212P

A Phase 1b/2 Study of Palazestrant (OP-1250) in Combination with Ribociclib in Patients with Estrogen Receptor-Positive, Human Epidermal Growth Factor Receptor 2-Negative, Advanced and/or Metastatic Breast Cancer

Virginia F. Borges[1], Jo Chien[2], Sara Nunnery[3], Jason A. Mouabbi[4], Cynthia Ma[5], Shakeela Bahadur[6], Jorge Chaves[7], David A. Potter[8], Dhanusha Sabanathan[9], Antoinette R. Tan[10], Lixian Sun[11], Demiana Faltaos[11], Margaret Tonda[11], Nancy U. Lin[12]

[1]University of Colorado Cancer Center, Aurora, CO, USA; [2]University of California San Francisco, San Francisco, CA, USA; [3]Vanderbilt University Medical Center, Nashville, TN, USA; [4]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [5]Washington University, St. Louis, MO, USA; [6]Banner MD Anderson Cancer Center, Gilbert, AZ, USA; [7]Northwest Medical Specialties, Tacoma, WA, USA; [8]University of Minnesota, Minneapolis, MN, USA; [9]Macquarie University, Sydney, NSW, Australia; [10]Levine Cancer Institute, Atrium Health, Charlotte, NC, USA; [11]Olema Oncology, San Francisco, CA, USA; [12]Dana-Farber Cancer Institute, Boston, MA, USA

INTRODUCTION

- Addition of cyclin-dependent kinase 4/6 (CDK4/6) inhibitors to endocrine therapy (ET) has improved patient outcomes for those with estrogen receptor-positive (ER+) advanced or metastatic breast cancer and is the current standard of care for first-line treatment.[1]
- Resistance to this first-line therapy eventually develops, most commonly due to activating mutations in the estrogen receptor 1 *(ESR1)* gene.[2] Suppression of both *ESR1*-wild type (*ESR1*-wt) and *ESR1*-mutant (*ESR1*-mut) estrogen receptor (ER) activity has the potential to significantly improve the current standard of care for first-line treatment.
- Palazestrant is a complete ER antagonist (CERAN) and selective ER degrader (SERD); it blocks AF1 and AF2 transcriptional activation domains of the ER.[3] (Figure 1)
- Palazestrant has demonstrated activity in both *ESR1*-wt and *ESR1*-mut preclinical models.[3]

Figure 1. Mechanism of action of palazestrant at the estrogen receptor[4]

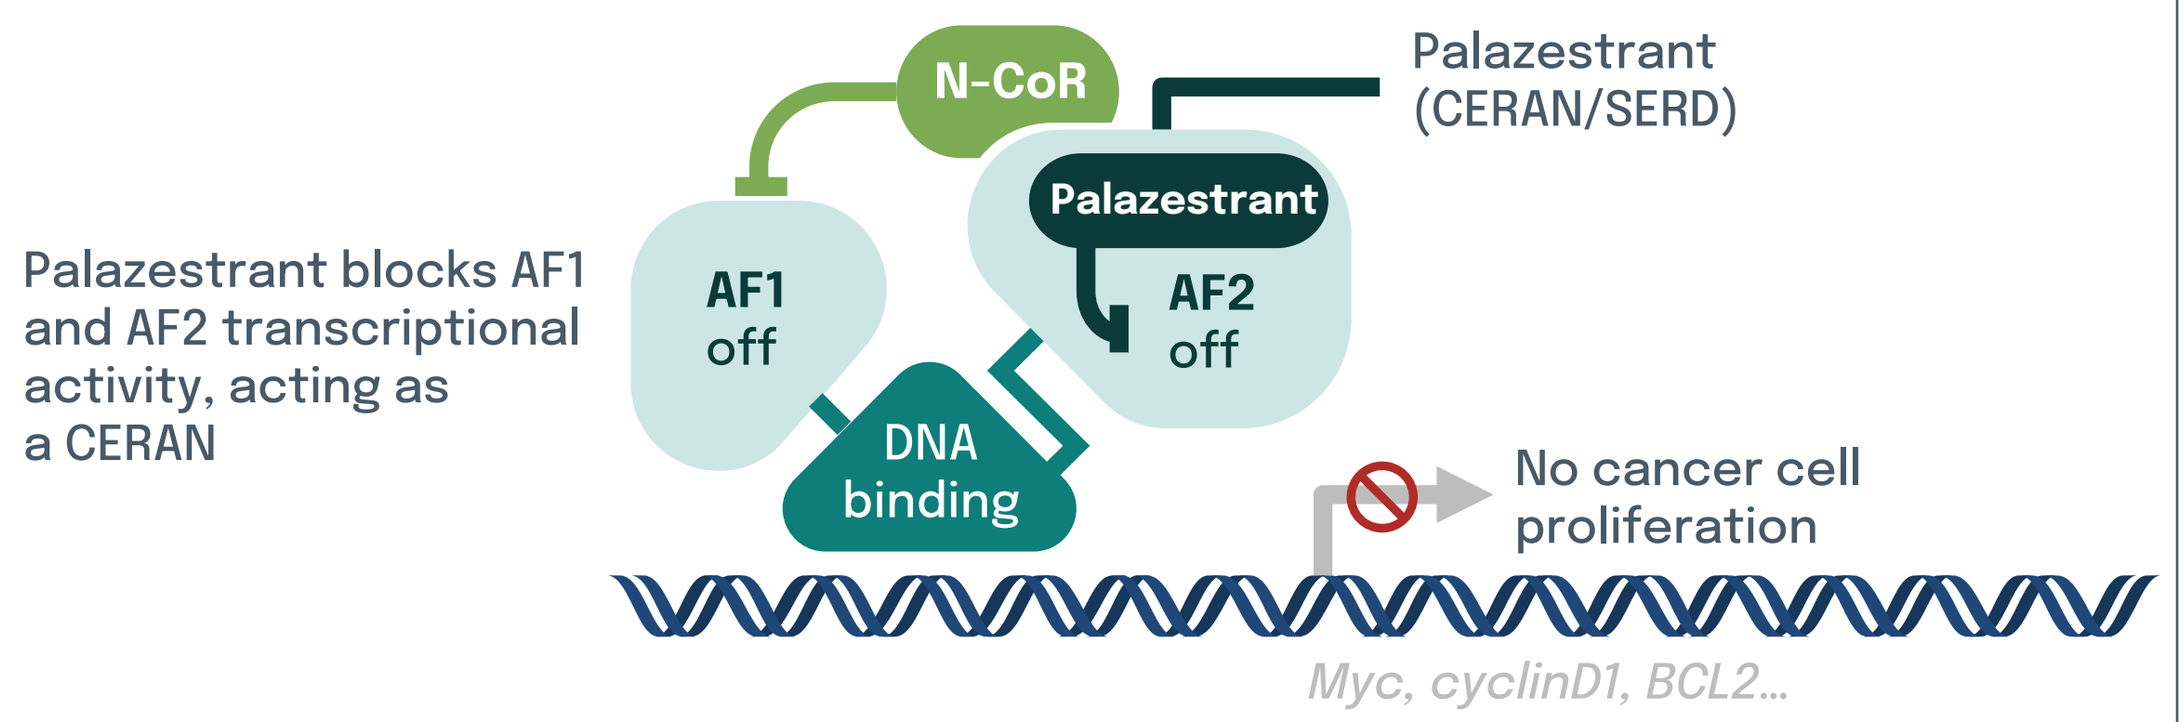
Palazestrant blocks AF1 and AF2 transcriptional activity, acting as a CERAN
N-CoR
Palazestrant (CERAN/SERD)
Palazestrant
AF1 off
AF2 off
DNA binding
No cancer cell proliferation
Myc, cyclinD1, BCL2...

AF, activation function; **CERAN,** complete estrogen receptor antagonist; **N-CoR,** nuclear receptor corepressor; **SERD,** selective estrogen receptor degrader.

- Palazestrant in combination with ribociclib resulted in enhanced tumor shrinkage and prolonged survival in both ERα wild type and mutant xenograft models.[5,6]
- In a phase 1/2 study, palazestrant as monotherapy demonstrated favorable safety, good tolerability, and a pharmacokinetics profile supportive of once-daily oral dosing in patients with ER+/human epidermal growth factor receptor 2-negative (HER2−) advanced or metastatic breast cancer. Encouraging tolerability and efficacy were observed.[7] A phase 3 study, OPERA-01, comparing palazestrant and SOC, is ongoing (NCT06016738).
- The recommended phase 2 dose (RP2D) of palazestrant as monotherapy or in combination with palbociclib or ribociclib is 120 mg QD.
- Here, we present an analysis from the OP-1250-003 study of palazestrant in combination with ribociclib in patients with ER+/HER2− advanced or metastatic breast cancer.

METHODS

- The OP-1250-003 study comprises a dose-escalation phase, followed by a dose-expansion phase. The study investigates palazestrant in combination with ribociclib, alpelisib, or everolimus. This analysis focuses on the palazestrant and ribociclib combination. (Figure 2)

Figure 2. OP-1250-003 study design: ribociclib group

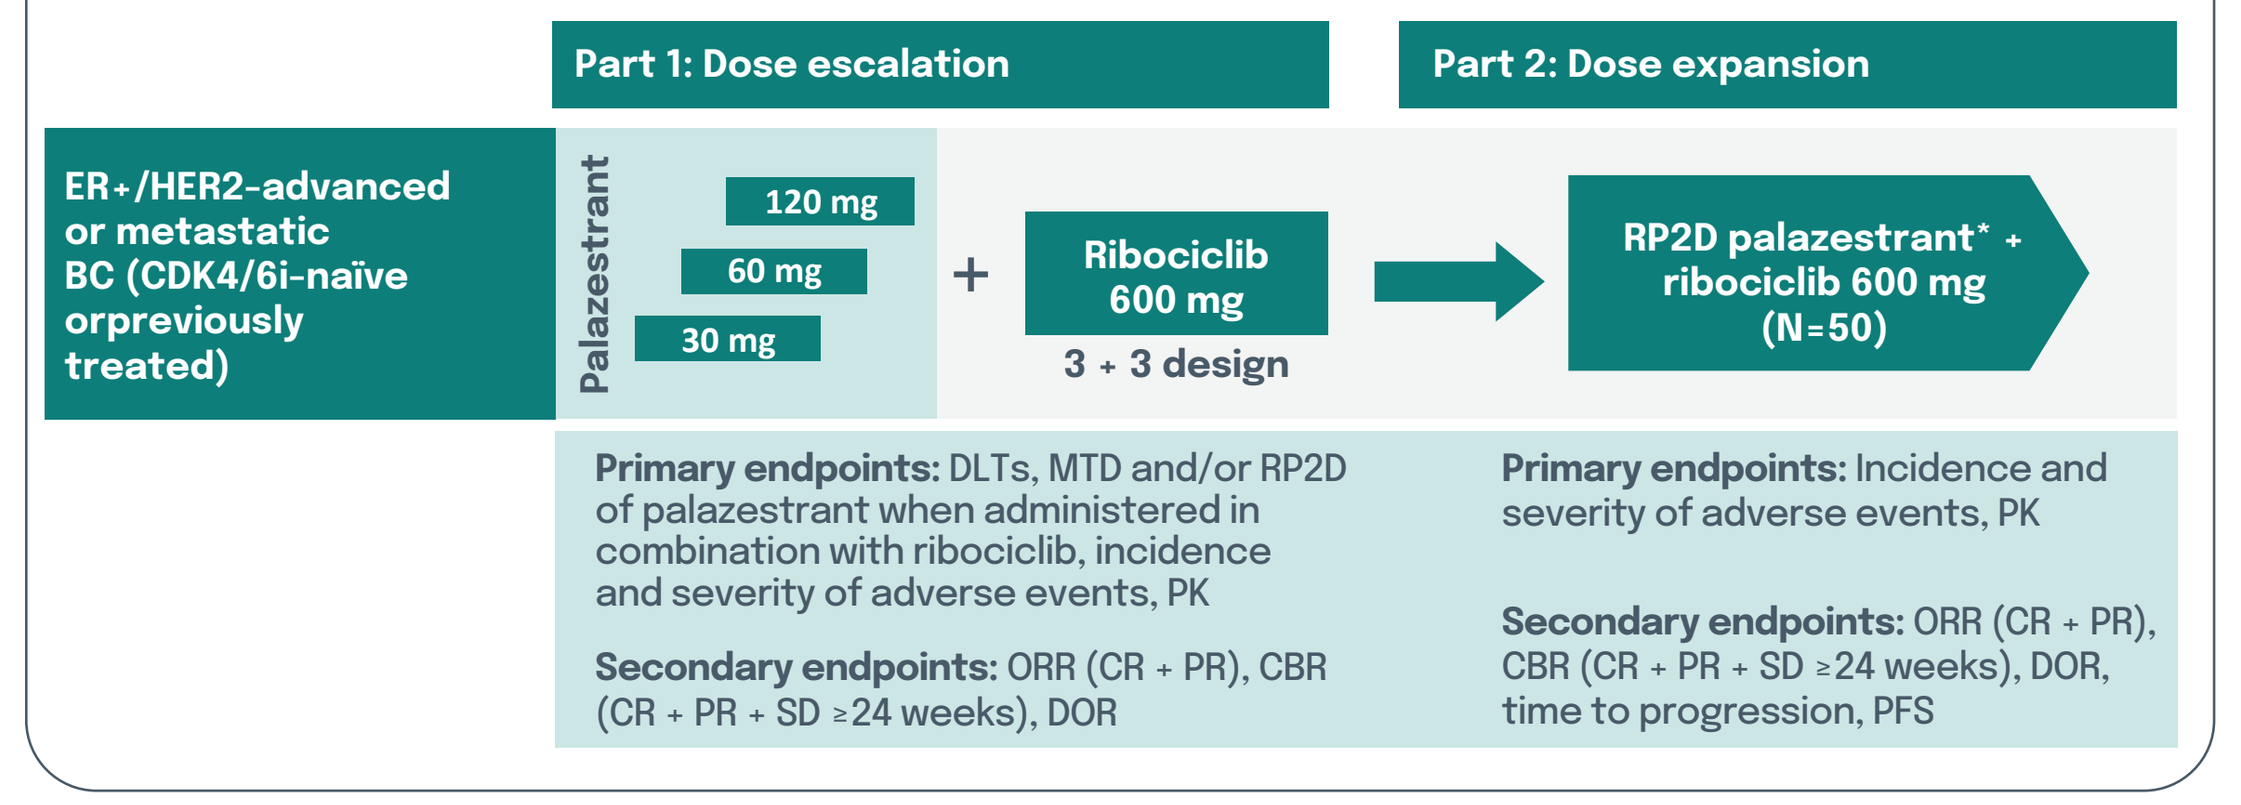
Part 1: Dose escalation
Part 2: Dose expansion
ER+/HER2-advanced or metastatic BC (CDK4/6i-naïve orpreviously treated)
Palazestrant
120 mg
60 mg
30 mg
+
Ribociclib 600 mg
3 + 3 design
RP2D palazestrant* + ribociclib 600 mg (N=50)
Primary endpoints: DLTs, MTD and/or RP2D of palazestrant when administered in combination with ribociclib, incidence and severity of adverse events, PK
Secondary endpoints: ORR (CR + PR), CBR (CR + PR + SD ≥24 weeks), DOR
Primary endpoints: Incidence and severity of adverse events, PK
Secondary endpoints: ORR (CR + PR), CBR (CR + PR + SD ≥24 weeks), DOR, time to progression, PFS

*Palazestrant was administered in capsules and in fasting state in this analysis.

BC, breast cancer; **CBR,** clinical benefit rate; **CDK4/6i,** cyclin-dependent kinase 4/6 inhibitor; **CR,** complete response; **DLT,** dose-limiting toxicity; **DOR,** duration of response; **ER+,** estrogen receptor-positive; **HER2,** human epidermal growth factor receptor 2; **MTD,** maximum tolerated dose; **ORR,** objective response rate; **PFS,** progression-free survival; **PK,** pharmacokinetics; **PR,** partial response; **RP2D,** recommended phase 2 dose; **SD,** stable disease.

PATIENT POPULATION

Key eligibility criteria

- Women or men with ER+/HER2− advanced or metastatic breast cancer.
- 0 to 2 prior endocrine therapies ± a CDK4/6 inhibitor for locally advanced or metastatic disease; 1 prior line of chemotherapy for advanced or metastatic breast cancer was allowed.
- Evaluable disease (measurable by RECIST 1.1 or bone only).
- Screening QTcF <450 ms, resting heart rate 50-90 beats/min.

DEMOGRAPHICS

- 60 patients were enrolled as of March 13, 2024; 50 patients who had 4 weeks or more of follow-up were included in this analysis. (Table 1)
- The dose-escalation part included 3 dose cohorts (3 patients per cohort): palazestrant at 30, 60, and 120 mg once daily plus ribociclib 600 mg once daily on days 1–21 of each 28-day treatment cycle (Figure 2). The RP2D of palazestrant was 120 mg in combination with ribociclib.
- 41 patients in the dose-expansion part (ongoing) received palazestrant 120 mg plus ribociclib 600 mg.
- 35 (70%) patients received prior treatment with CDK4/6 inhibitors in combination with endocrine therapy for advanced disease. (Table 1)
 - 24 (48%) patients received 1 prior treatment with a CDK4/6 inhibitor (palbociclib, n=20; abemaciclib n=3; ribociclib, n=1).
 - 11 (22%) patients received 2 prior treatments with CDK4/6 inhibitors (palbociclib→palbociclib, n=3; palbociclib→abemaciclib, n=3; palbociclib→ribociclib, n=3; ribociclib→ribociclib, n=1; palbociclib→an experimental CDK4/6 inhibitor, n=1).

Table 1. Baseline patient demographics and disease characteristics

Characteristic	Total (N=50)
Median age, years (range)	62 (37–85)
Female sex	50 (100%)
Premenopausal	6 (12%)
ECOG Performance Status	
0	24 (48%)
1	22 (44%)
Not reported	4 (8%)
Measurable disease	31 (62%)
Visceral disease	29 (58%)
Prior lines of therapy for advanced disease	
0	13 (26%)
1	20 (40%)
2	12 (24%)
3	5 (10%)
Prior lines of endocrine therapy for advanced disease	
0	13 (26%)
1	24 (48%)
2	13 (26%)
Types of prior therapy for advanced disease	
CDK4/6 inhibitor	35 (70%)
Aromatase inhibitor	29 (58%)
Fulvestrant	18 (36%)
Chemotherapy	9 (18%)
ESR1 mutation at baseline (ctDNA)	13/48* (27%)

**ESR1* mutations in ctDNA at baseline were determined centrally using SafeSEQ Breast Cancer Panel (Sysmex Inostics, Baltimore, MD). Two samples were not evaluable.
Data cutoff: March 13, 2024.
CDK4/6, cyclin-dependent kinase 4/6; **ctDNA,** circulating tumor DNA; **ECOG,** Eastern Cooperative Oncology Group; ***ESR1*,** estrogen receptor 1 gene.

SAFETY AND TOLERABILITY

- No dose-limiting toxicities (DLTs) were observed during the dose-escalation phase (the DLT observation period was at least 4 weeks), and the maximum tolerated dose was not reached.
- The majority of treatment-emergent adverse events (TEAEs) were Grade 1 and Grade 2. (Table 2)
- No patients discontinued palazestrant due to a treatment-related adverse event (AE). (Table 3)
- 2 patients discontinued ribociclib without discontinuation of palazestrant in the 120 mg cohort for neutropenia (Grade 3, n=1; Grade 4, n=1).
- 3 patients had dose reduction of palazestrant and ribociclib due to neutropenia (Grade 3, n=2; Grade 2, n=1).
- No patients had a dose reduction of only palazestrant.
- 10 patients had dose reduction of ribociclib only. Reasons for dose reduction were QTcF prolongation (n=4; Grade 3, n=2; Grade 2, n=1; Grade 1, n=1), neutropenia (n=4; Grade 3, n=3; Grade 2, n=1) or fatigue (n=2; Grade 2).
- Neutropenia was reversible in all patients, and the timing was consistent with the ribociclib-related neutropenia.[8,9]

Table 2. Treatment-emergent adverse events (TEAEs) reported in ≥20% of patients

TEAEs ≥20%	Olema Study 003 Ribociclib + Palazestrant (n=50)			MONALEESA-2 Ribociclib + Letrozole* (n=334)		
	All grades†	Grade 3	Grade 4	All grades	Grade 3	Grade 4
Neutropenia‡	38 (76%)	24 (48%)	5 (10%)	93%§,¶	49%	11%
Nausea	37 (74%)	1 (2%)	0	52%	2%	0
Fatigue	25 (50%)	1 (2%)	0	37%	2%	<1%
Diarrhea	23 (46%)	1 (2%)	0	35%	1%	0
Anemia	18 (36%)	1 (2%)	0	57%¶	2%	0
WBC count decreased	18 (36%)	8 (16%)	0	93%¶	31%	3%
Constipation	15 (30%)	0	0	25%	1%	0
Creatinine increased	12 (24%)	0	0	20%¶	1%	0
Electrocardiogram QT prolonged	12 (24%)	3 (6%)	0	43%#	8%#	0
Thrombocytopenia	10 (20%)	0	0	29%¶	1%	0

Data shown are n or n (%). *Adverse reactions reported in ≥10% of patients. Source: Novartis Kisqali (ribociclib) Prescribing Information, 2022. †2 patients experienced Grade 5 AEs (myocarditis due to COVID-19; depressed level of consciousness not related to study drug). ‡Combined term includes neutropenia and decreased neutrophil count. §Reported as neutrophil count or leukocyte count decreased in the laboratory abnormalities in the MONALEESA-2 study. ¶These values were taken from MONALEESA-2 lab abnormalities data. #Ribociclib + fulvestrant in MONALEESA-3 per the Ribociclib Approval Package (NUMBER:209092Orig1s001, June 2018); ribociclib + non-steroidal aromatase inhibitors in MONALEESA-7: all grade QTcF prolongation was 46.5% (Grade 2, 5.3%; Grade 3, 9%).
NR, not reported; **WBC,** white blood cell.

Table 3. Patient disposition

Treatment status	Palazestrant dose 30 mg (n=3)	60 mg (n=3)	120 mg (n=44)	Total (N=50)
Treatment ongoing	0	1	32	33 (66%)
Treatment discontinued	3	2	12	17 (34%)
Reasons for treatment discontinuation				
Disease progression	1	1	5	7 (14%)
Clinical progression	2	0	4	6 (12%)
Patient decision	0	0	2	2 (4%)
Other	0	0	1	1 (2%)
Adverse event	0	1*	0	1 (2%)

*Unrelated to study treatment, patient experienced myocarditis due to COVID-19.

PHARMACOKINETICS

- Palazestrant did not affect ribociclib 600 mg exposure when compared with published concentrations for single-agent ribociclib. (Figure 3)
- Steady-state trough values between the combination and single-agent palazestrant were overlapping. (Figure 4)

Figure 3. Steady-state exposure of ribociclib* (600 mg) in combination with palazestrant

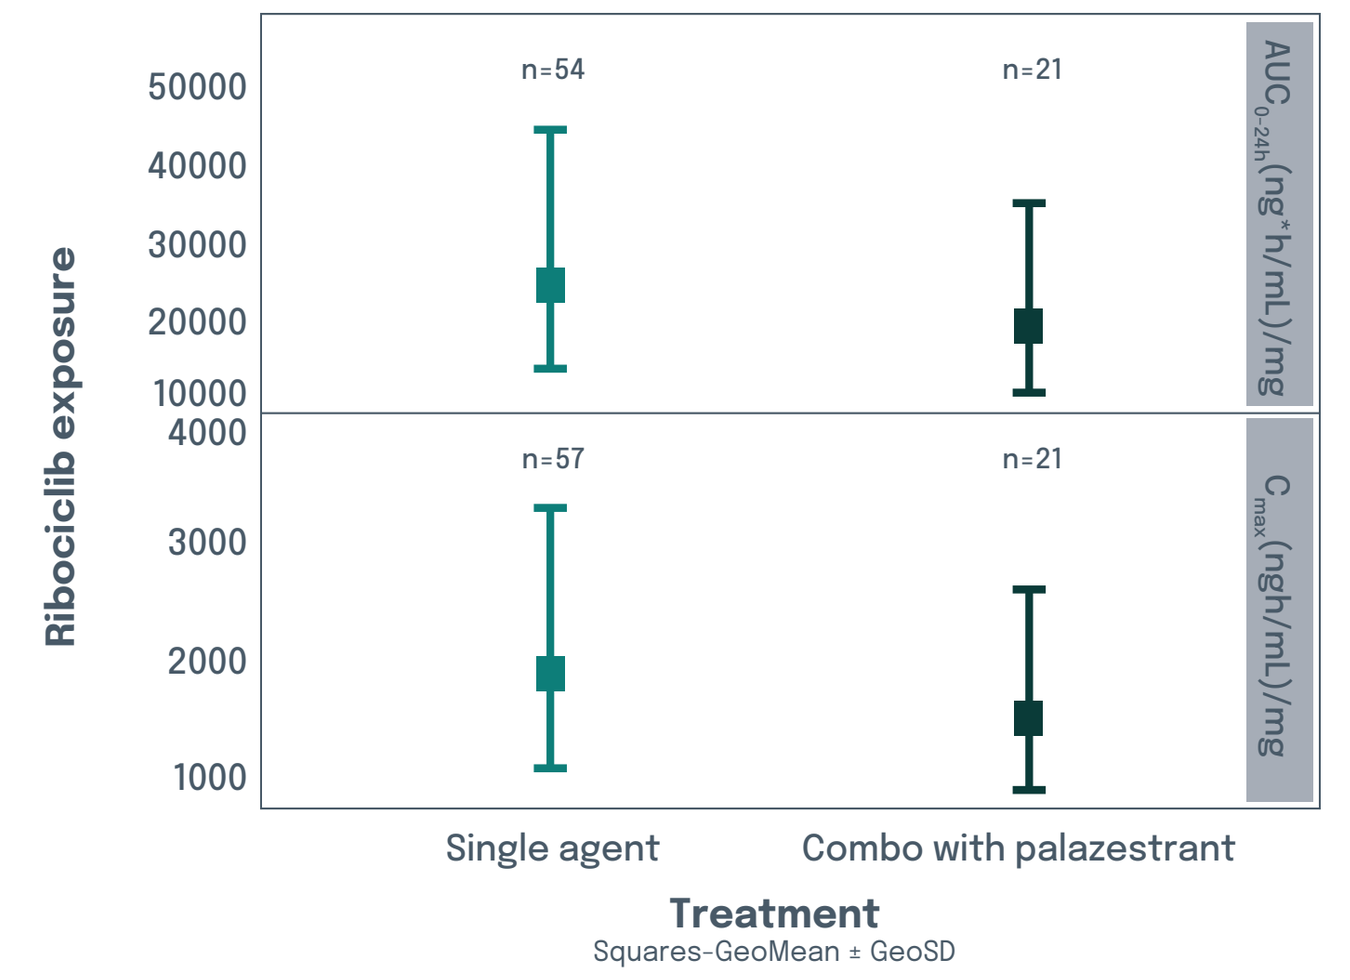
Ribociclib exposure
50000
40000
30000
20000
10000
4000
3000
2000
1000
n=54
n=21
n=57
n=21
AUC0-24h(ng*h/mL)/mg
Cmax(ngh/mL)/mg
Single agent
Combo with palazestrant
Treatment
Squares-GeoMean ± GeoSD

Data are geometric mean ± geometric standard deviation.
*Single-agent steady-state exposure levels for ribociclib.[10]
Data cutoff: March 20, 2024.
AUC_{0-24}, area under the curve from 0 to 24 h; **C_{max},** maximum concentration; **Combo**, combination; **GeoMean,** geometric mean; **GeoSD,** geometric standard deviation.

Figure 4. Palazestrant steady-state trough concentrations

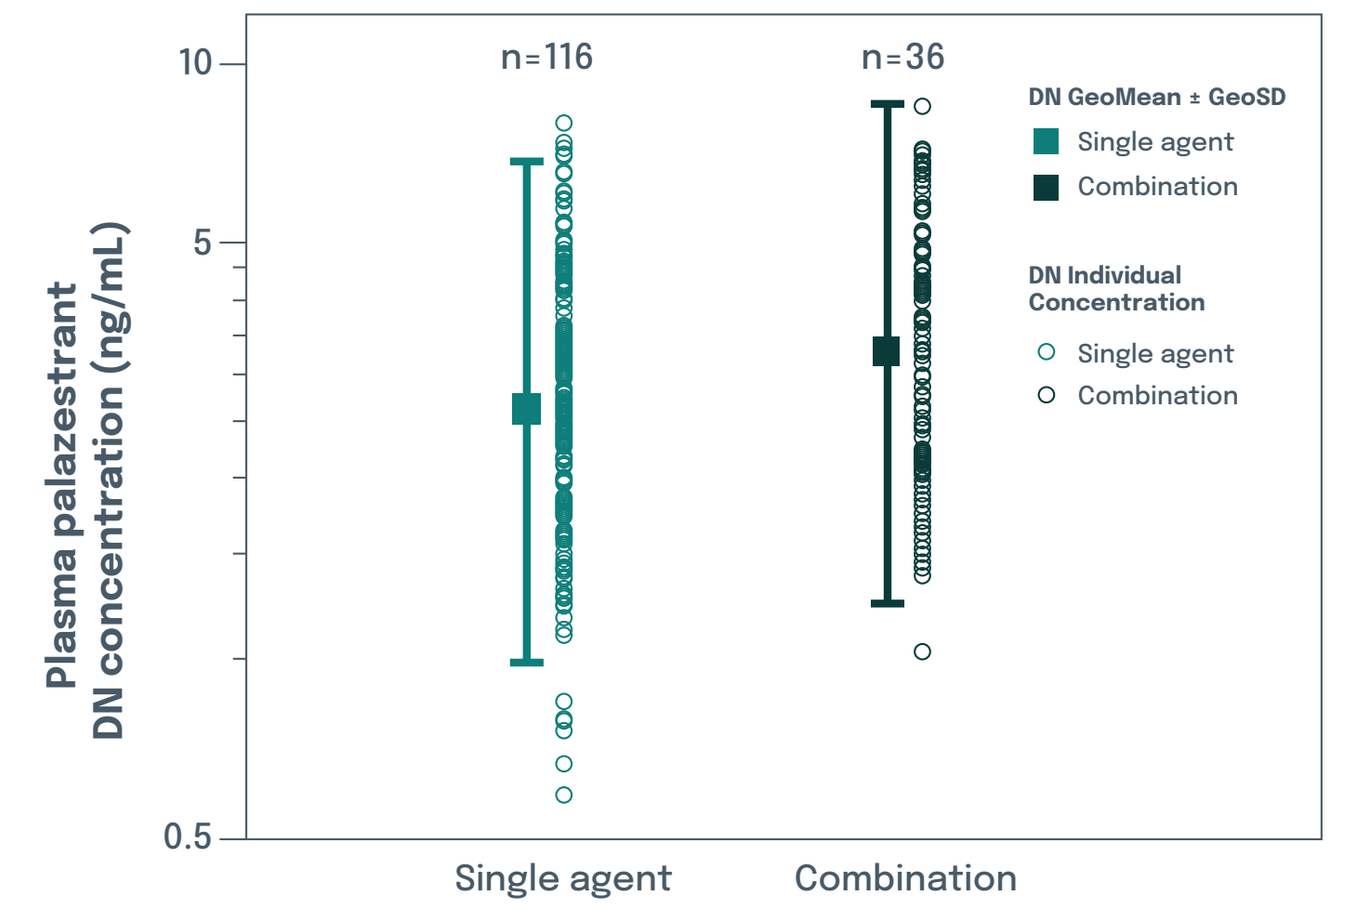
Plasma palazestrant DN concentration (ng/mL)
10
5
0.5
n=116
n=36
DN GeoMean ± GeoSD
Single agent
Combination
DN Individual Concentration
Single agent
Combination
Single agent
Combination

Note: Predose samples at C2D1, C2D15, C3D1, C5D1, C7D1, and C9D1 for both studies.
Data cutoff: March 20, 2024.
DN, dose normalized; **GeoMean,** geometric mean; **GeoSD,** geometric standard deviation.

EFFICACY

Figure 5. Duration of treatment*

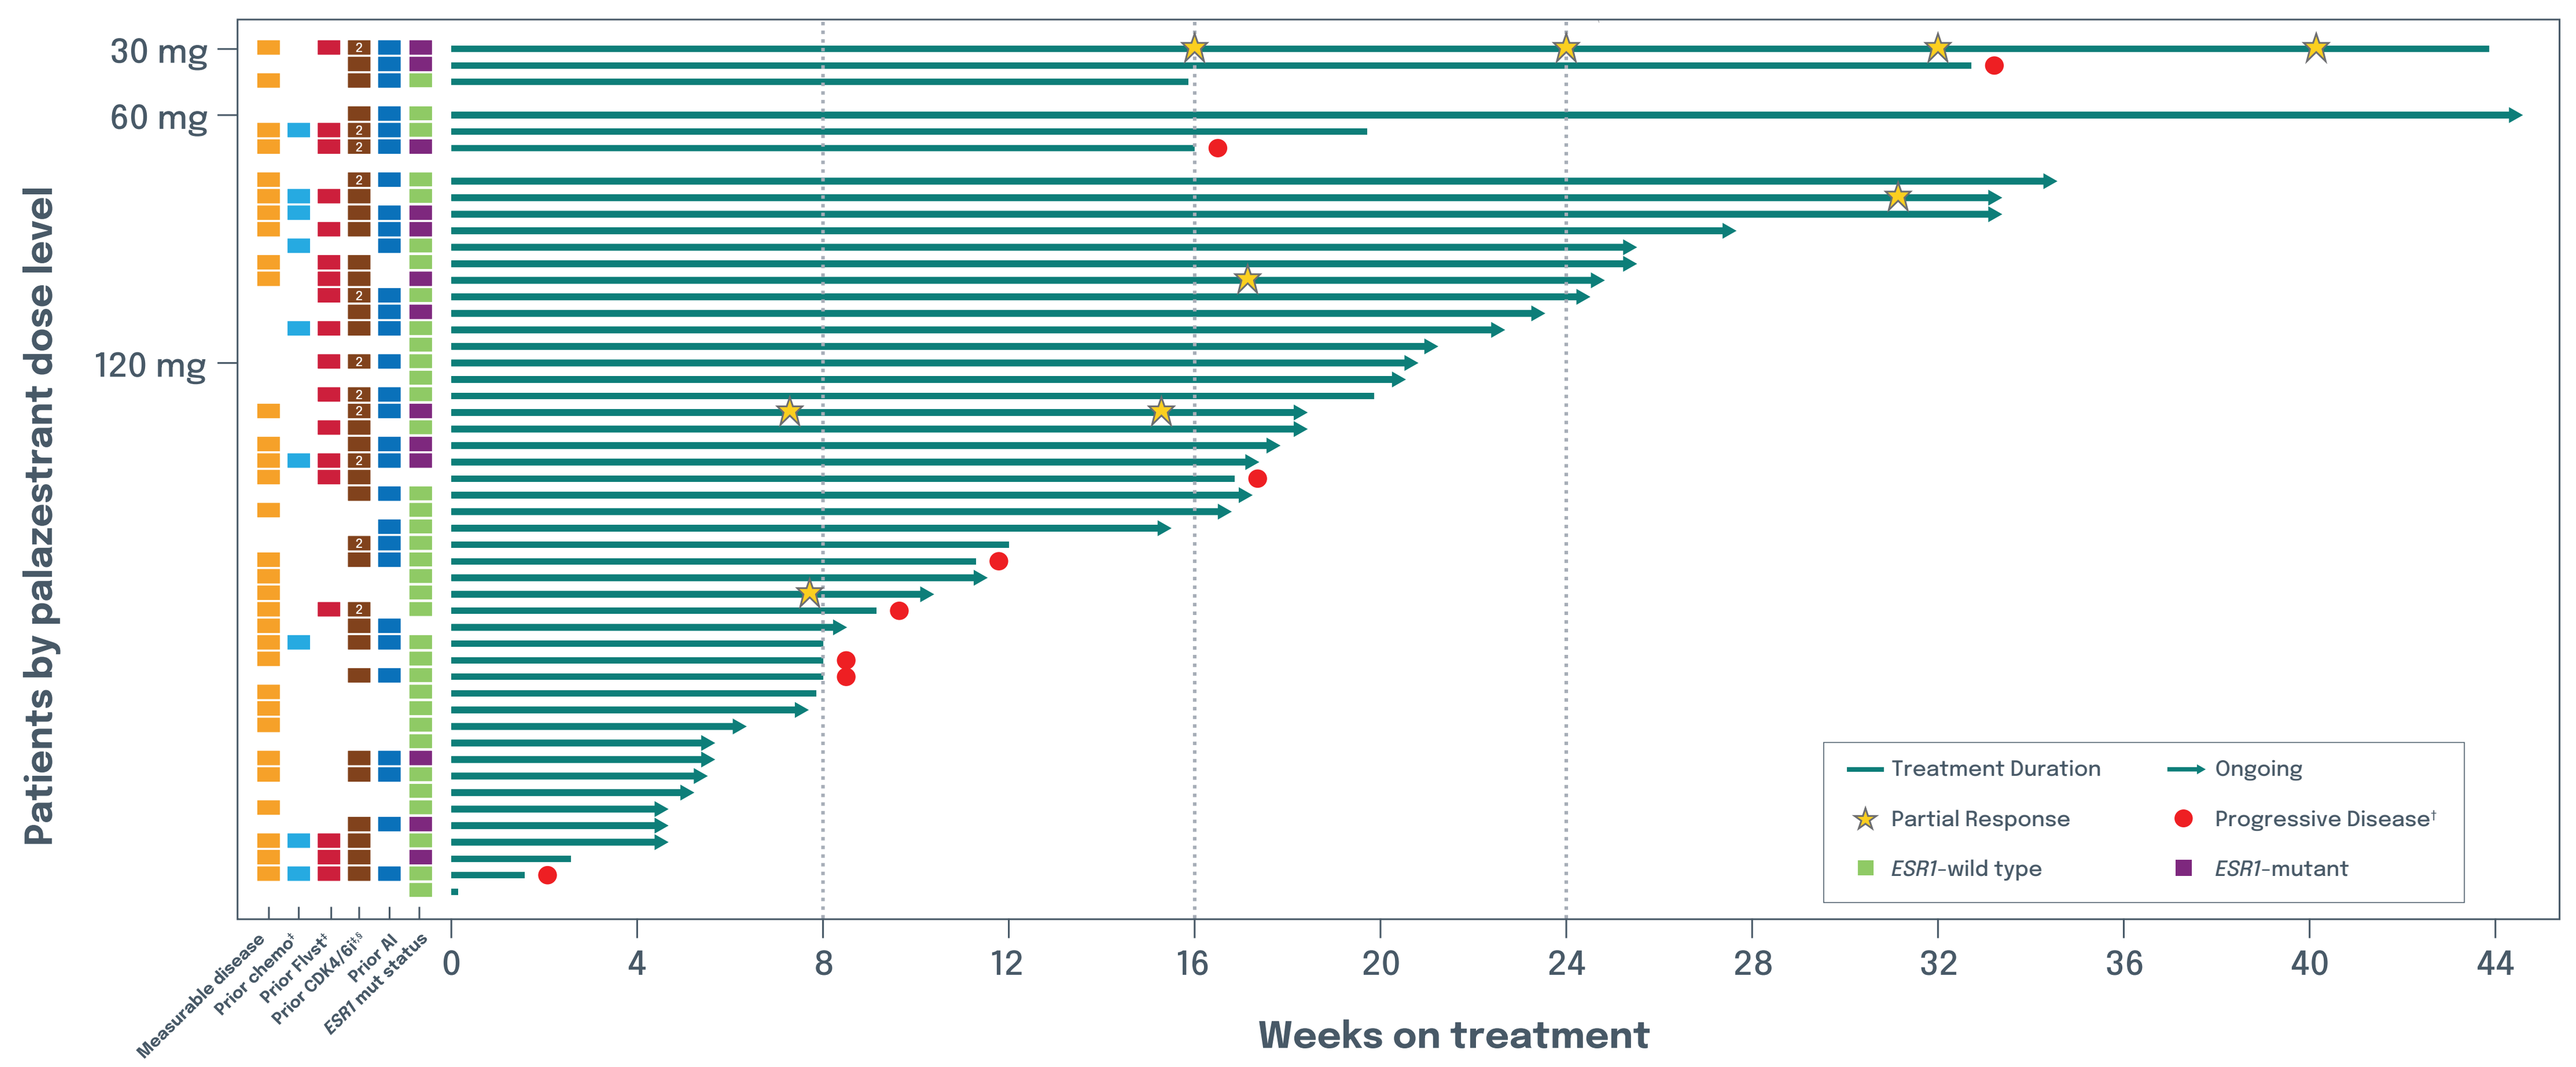
Patients by palazestrant dose level
30 mg
60 mg
120 mg
Measurable disease
Prior chemo‡
Prior Flvst‡
Prior CDK4/6i‡§
Prior AI
ESR1 mut status
0
4
8
12
16
20
24
28
32
36
40
44
Weeks on treatment
Treatment Duration
Ongoing
Partial Response
Progressive Disease†
ESR1-wild type
ESR1-mutant

*Each lane represents 1 patient. PD assessment date was used if end of treatment date was missing for patients with PD. †Progression by radiographic assessment only. ‡Prior therapies were in the advanced setting. §Patients who received 2 prior CDK4/6i in the metastatic setting are indicated with a 2.
AI, aromatase inhibitor; **CDK4/6i,** cyclin-dependent kinase 4/6 inhibitor; **chemo,** chemotherapy; ***ESR1*,** estrogen receptor 1 gene; **Flvst,** fulvestrant; **mut**, mutation; **PD,** progressive disease.

Figure 6. Best percent change from baseline (BL) in target lesions and best overall response*

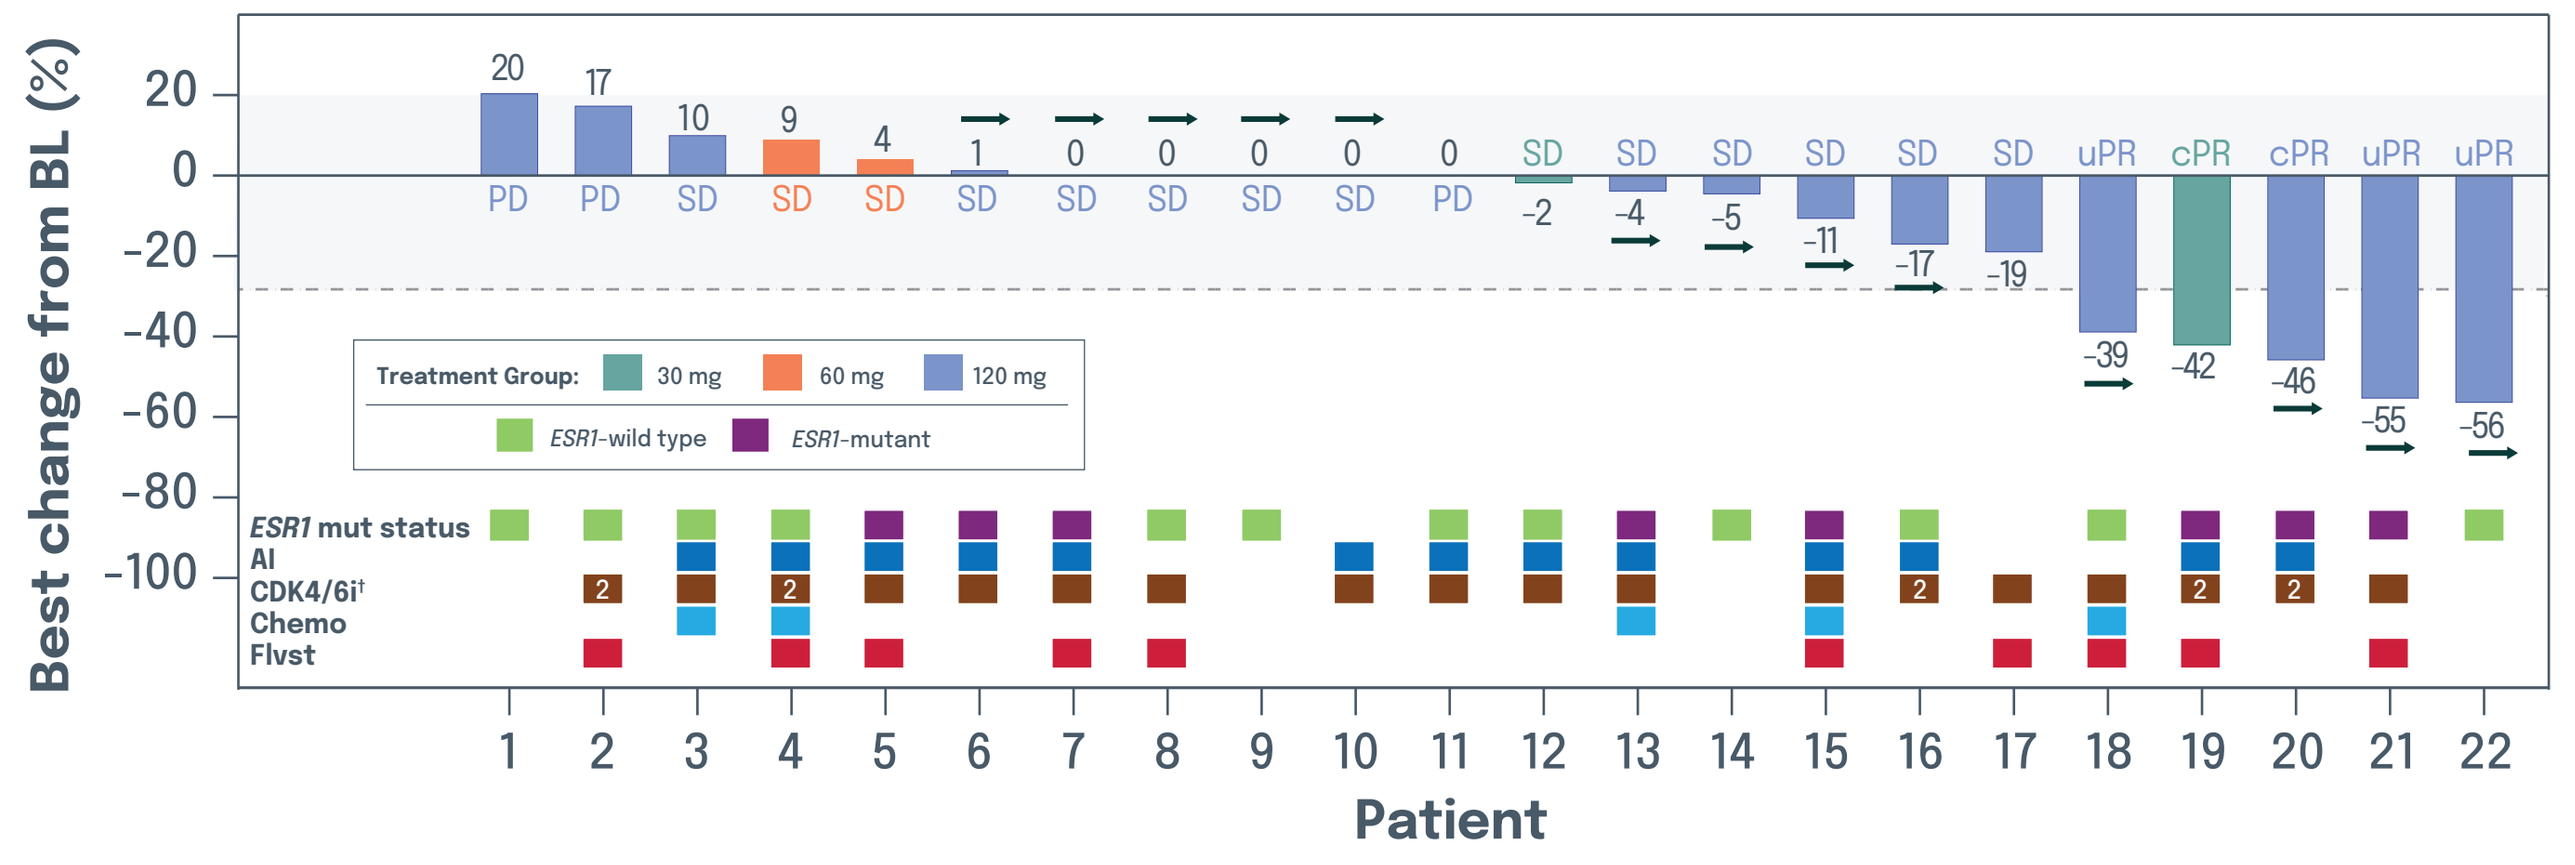
Best change from BL (%)
20
0
-20
-40
-60
-80
-100
20 17 10 9 4 1 0 0 0 0 0 -2 -4 -5 -11 -17 -19 -39 -42 -46 -55 -56
PD PD SD SD SD SD SD SD SD SD PD SD SD SD SD SD SD uPR cPR cPR uPR uPR
Treatment Group: 30 mg 60 mg 120 mg
ESR1-wild type ESR1-mutant
ESR1 mut status
AI
CDK4/6i†
Chemo
Flvst
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22
Patient

*Waterfall plot n=22. †Patients who received 2 prior CDK4/6i in the metastatic setting are indicated with a 2.
AI, aromatase inhibitor; **CDK4/6i,** cyclin-dependent kinase 4/6 inhibitor; **chemo,** chemotherapy; **cPR,** confirmed partial response; ***ESR1*,** estrogen receptor 1 gene; **Flvst,** fulvestrant; **mut**, mutation; **PD,** progressive disease; **SD,** stable disease; **uPR,** unconfirmed partial response.

- Partial responses were observed in 5 patients (2 cPR; 3 uPR in 5/23) to date.
- CBR was 85% (11/13 CBR-eligible*) in all patients.
 - 83% (5/6 CBR-eligible*) in patients with *ESR1* mutation detected.
 - 86% (6/7 CBR-eligible*) in patients with *ESR1* mutation not detected.
 - 83% (10/12 CBR-eligible*) in CDK4/6i-pretreated patients.
- 11 (50%) patients had reduction in target lesion size. (Figure 6)
- Longest duration of treatment is 44 weeks and ongoing. Of 50 patients, 33 (66%) remain on treatment. (Figure 5)

*CBR-eligible patients were patients who enrolled ≥24 weeks before data cutoff, received ≥1 cycle of palazestrant treatment, and had post-baseline tumor assessment.

CONCLUSIONS

The combination of palazestrant at 120 mg with ribociclib 600 mg was well tolerated; no new safety signals or enhancement of toxicity were identified.

- Severity and incidence of adverse events were consistent with the expected safety profile of ribociclib plus endocrine therapy and were similar to those reported previously.[8]

Palazestrant did not affect ribociclib exposure and ribociclib has no clinically meaningful effect on palazestrant exposure.

Promising efficacy was observed, including in patients who received prior treatment with CDK4/6 inhibitors.

- 5 PRs (2 confirmed + 3 unconfirmed) among 23 response-evaluable patients.
- CBR 85% among CBR-eligible patients: 83% in *ESR1*-mut and 86% in *ESR1*-wt.

Findings from this study support further clinical development of palazestrant in combination with CDK4/6 inhibitors for the first-line treatment of ER+/HER2− advanced or metastatic breast cancer.

References

1. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Breast Cancer. V2.2024. 2. Rasha F, et al. *Mol Cell Endocrinol.* 2021;532:111322. 3. Alemany C, et al. Presented at AACR-NCI-EORTC; October 7-10, 2021 (abstract number P037). 4. Shang Y, Brown M. *Science.* 2002;295:2465–8. 5. Webb P, et al. *J Biol Chem.* 2003;278:6912–20. 6. Parisian AD, et al. Presented at SABCS 2022; December 6-10, 2022 (poster number P2-24-07). 7. Lin NU, et al. Presented at ESMO Congress; October 20-24, 2023 (presentation number 383MO). 8. KISQALI (ribociclib). Prescribing information. Novartis; 2022. 9. Thill M, Schmidt M. *Ther Adv Med Oncol.* 2018; 10:1-12. 10. Yan J, et al. Presented at SABCS 2019; December 10-14, 2019 (poster number P1-19-37).

Acknowledgments

We thank the OP-1250-003 trial patients, their families and caregivers, trial investigators, and study staff. This study was sponsored by Olema Oncology. Editorial support was provided by Anuradha Kumari, PhD, and Melanie Styers, PhD, of Veracity Science, LLC, and funded by Olema Oncology.

Disclosures

Dr. Borges reports consulting fees from Seagen, AstraZeneca, and Gilead. She is the lead site investigator on clinical trials sponsored by Olema Oncology, AstraZeneca, and Seagen, and her institution receives clinical trial fees from these companies.

Sponsored by Olema Oncology

Presented at ESMO Breast Cancer 2024; Berlin, Germany; May 15–17, 2024