A Phase 1b/2 Study of OP-1250, an Oral Complete Estrogen Receptor Antagonist (CERAN) and Selective ER Degrader (SERD) with Palbociclib in Patients with Advanced or Metastatic HR+/HER2– Breast Cancer

202P

Arlene Chan,[1] Phuong Dinh,[2] Daphne Day,[3] Michael Slancar,[4] Vinod Ganju,[5] Nicole McCarthy,[6] Janine Lombard,[7] Demiana Faltaos,[8] Mark Shilkrut,[8] Rosalind Wilson,[8] Caitlin Murphy[9]

[1]Breast Cancer Research Centre-WA, Curtin University, Breast Clinical Trials Unit, Hollywood Private Hospital, Nedlands, WA, Australia; [2]Crown Princess Mary Cancer Care Centre, Westmead, NSW, Australia; [3]Monash Health, Monash University, Clayton, VIC, Australia; [4]ICON Cancer Centre, Southport, QLD, Australia; [5]Peninsula & South Eastern Haematology and Oncology Group, Frankston, VIC, Australia; [6]ICON Cancer Centre Wesley, Auchenflower, QLD, Australia; [7]Calvary Mater Newcastle, Waratah, NSW, Australia; [8]Olema Oncology, San Francisco, CA, USA; [9]Cancer Services Trials Unit, University Hospital Geelong, Barwon Health, Geelong, VIC, Australia

Introduction

- Addition of cyclin-dependent kinase 4/6 (CDK4/6) inhibitors to endocrine therapy has improved outcomes in patients with hormone receptor-positive (HR+) advanced or metastatic breast cancer (MBC) and is the current standard of care for first-line treatment[1]
- Resistance to this first-line treatment eventually develops with mutations in estrogen receptor 1 *(ESR1)* constituting the most common mechanism.[2] The ability to suppress the activity of both wild-type and *ESR1* mutant estrogen receptors (ER) represents the potential to significantly improve upon current standard of care
- OP-1250 is a complete ER antagonist (CERAN) and selective ER degrader (SERD), which blocks both the AF1 and AF2 transcriptional activation domains of the ER[3] (**Figure 1**). OP-1250 has demonstrated activity in both *ESR1* wild-type and *ESR1* mutant pre-clinical models[3]

Figure 1. Mechanism of action of OP-1250 at the estrogen receptor[4,5]

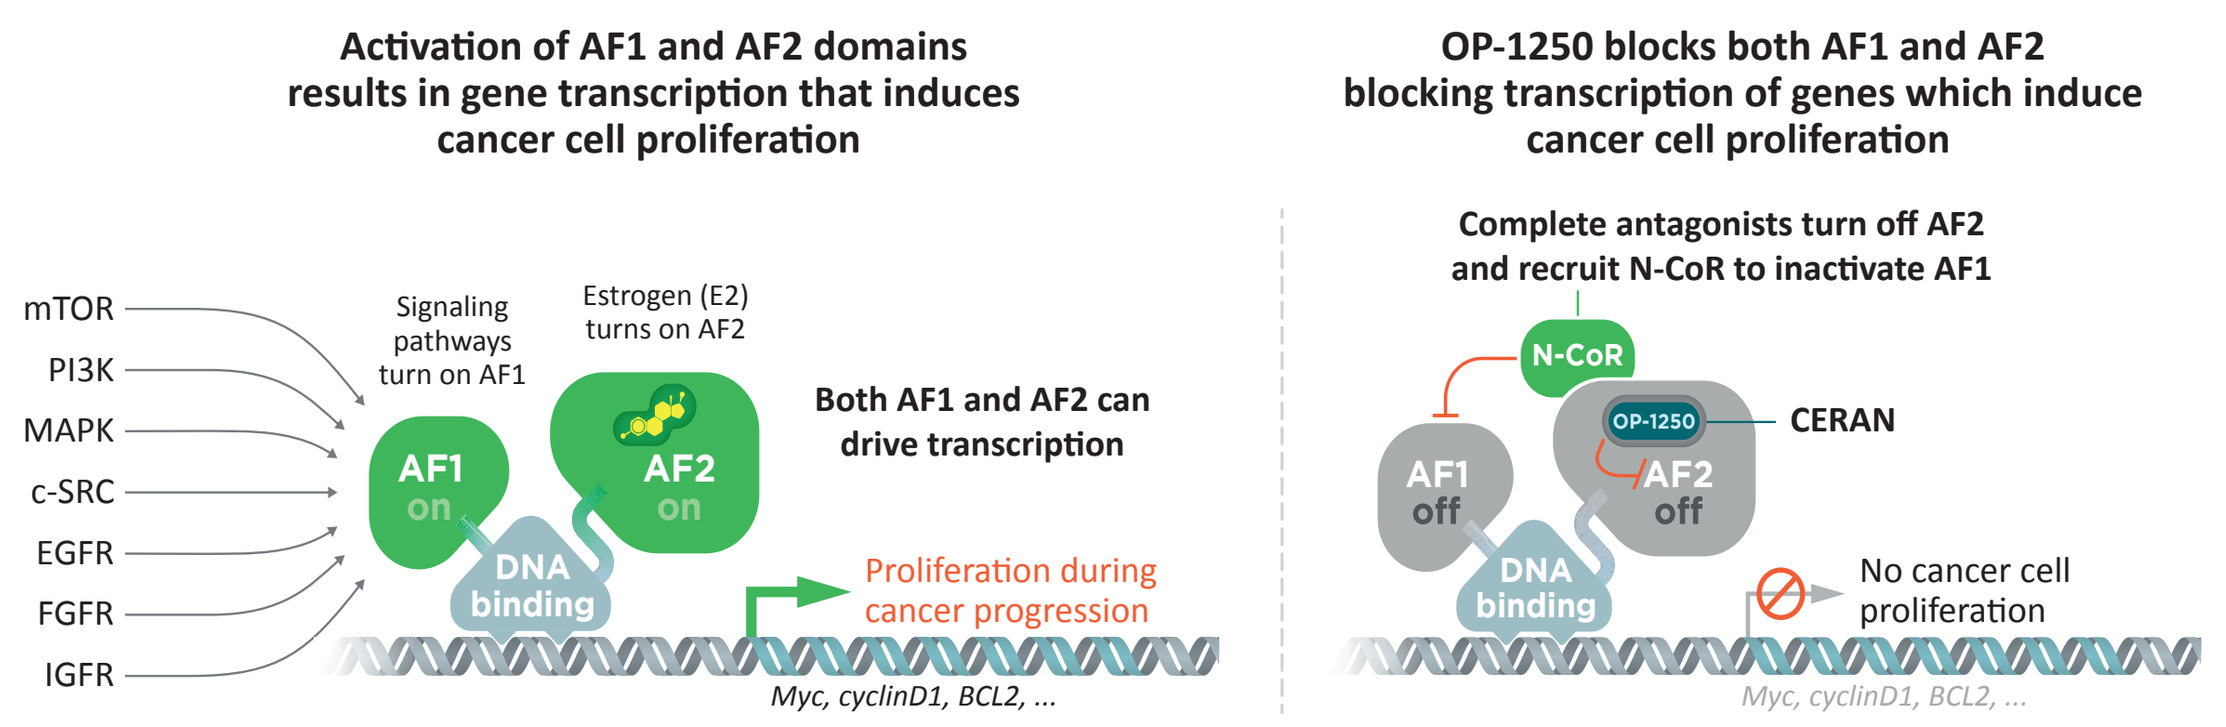
Activation of AF1 and AF2 domains results in gene transcription that induces cancer cell proliferation
OP-1250 blocks both AF1 and AF2 blocking transcription of genes which induce cancer cell proliferation
mTOR
PI3K
MAPK
c-SRC
EGFR
FGFR
IGFR
Signaling pathways turn on AF1
Estrogen (E2) turns on AF2
AF1 on
AF2 on
DNA binding
Both AF1 and AF2 can drive transcription
Proliferation during cancer progression
Myc, cyclinD1, BCL2, ...
Complete antagonists turn off AF2 and recruit N-CoR to inactivate AF1
N-CoR
OP-1250
CERAN
AF1 off
AF2 off
No cancer cell proliferation

AF, activation function; **CERAN**, complete estrogen receptor antagonist; **N-CoR**, nuclear receptor corepressor.

- Results from an ongoing phase 1/2 study with OP-1250 as monotherapy (NCT04505826) demonstrated acceptable safety, good tolerability, and a pharmacokinetics profile supportive of once-daily oral dosing in patients with HR+/human epidermal growth factor receptor 2-negative (HR+/HER2–) advanced or MBC. Objective tumor responses and prolonged disease stabilizations were demonstrated in these heavily pretreated patients[6]
- Previous data from the open-label, phase 1/2 study of OP-1250 in combination with the CDK4/6 inhibitor palbociclib in patients with HR+/HER2– MBC (NCT05266105) has established the recommended phase 2 dose.[7] Here we report an updated pharmacokinetics profile, drug-drug interaction (DDI), safety, and available antitumor activity data from this ongoing study

Methods

- This two-part study is comprised of a dose-escalation phase followed by a dose-expansion phase (**Figure 2**)

Figure 2. OP-1250-002 study design

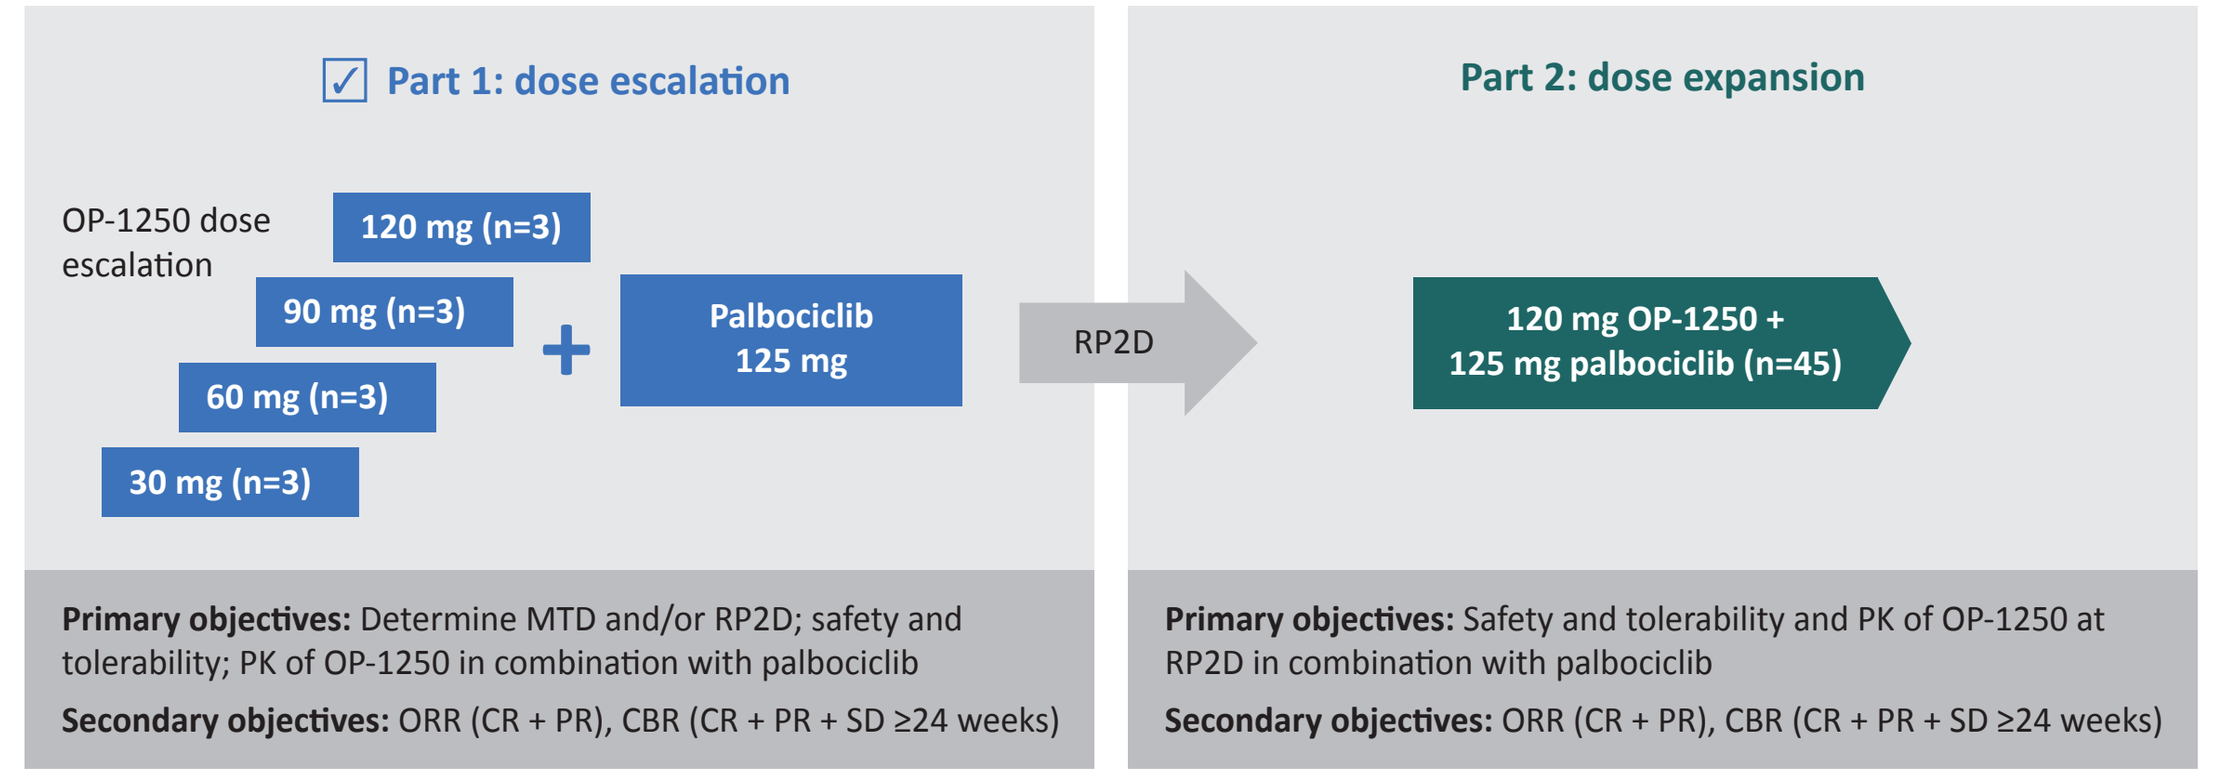
Part 1: dose escalation
OP-1250 dose escalation
120 mg (n=3)
90 mg (n=3)
60 mg (n=3)
30 mg (n=3)
Palbociclib 125 mg
RP2D
Part 2: dose expansion
120 mg OP-1250 + 125 mg palbociclib (n=45)
Primary objectives: Determine MTD and/or RP2D; safety and tolerability; PK of OP-1250 in combination with palbociclib
Secondary objectives: ORR (CR + PR), CBR (CR + PR + SD ≥24 weeks)
Primary objectives: Safety and tolerability and PK of OP-1250 at RP2D in combination with palbociclib
Secondary objectives: ORR (CR + PR), CBR (CR + PR + SD ≥24 weeks)

CBR, clinical benefit rate; **CR**, complete response; **MTD**, maximum tolerated dose; **ORR**, overall response rate; **PK**, pharmacokinetic; **PR**, partial response; **RP2D**, recommended phase 2 dose; **SD**, stable disease.

- Key eligibility criteria
 - Women (regardless of menopausal status) or men with HR+/ HER2– advanced BC or MBC
 - Zero or one prior endocrine therapy ± a CDK4/6 inhibitor for locally advanced or metastatic disease. One prior line of chemotherapy for advanced or MBC was allowed
 - Evaluable disease (measurable or non-measurable by RECIST 1.1)

Patient Population

- 29 patients were enrolled as of March 8, 2023 (**Table 1**)
 - The dose-escalation part included four dose cohorts (three patients per cohort): OP-1250 at 30, 60, 90, and 120 mg once daily plus palbociclib 125 mg once daily on days 1–21 of each 28-day treatment cycle (**Table 2**)
 - In the dose-expansion part (ongoing), patients receive OP-1250 120 mg plus palbociclib 125 mg. Seventeen patients had been enrolled by the data cutoff date, with total planned enrollment of ~45 patients
- 24 (83%) patients received prior endocrine therapy for advanced disease
- 20 (69%) patients received prior treatment with CDK4/6 inhibitors for advanced disease (palbociclib, n=14; ribociclib, n=5; both palbociclib and ribociclib, n=1)

Table 1. Baseline patient demographics and disease characteristics

Baseline characteristics	Total (N=29)
Median age, years (range)	65 (46–76)
Female	28 (97)
Premenopausal	2 (7)
ECOG performance status	
0	19 (66)
1	10 (35)
Measurable disease at baseline	23 (79)
Visceral disease (liver, lung, peritoneum, pleura, ascites)	15 (52)
Prior lines of therapy for advanced disease	
0	2 (7)
1	20 (69)
2	7 (24)[a]
Prior lines of endocrine therapy for advanced disease	
0	5 (17)
1	24 (83)
Types of prior therapy for advanced disease	
CDK4/6 inhibitor	20 (69)
Aromatase inhibitor	21 (72)
Fulvestrant	3 (10)
Chemotherapy	6 (21)
ESR1 mutations at baseline (ctDNA)	8/18 (44)[b]

Data shown are n (%) or n/N (%) unless otherwise specified. [a]One patient received chemotherapy, endocrine therapy, and olaparib. [b]ctDNA from 18 patients was available at the time of data cutoff for *ESR1* mutation evaluation.
CDK, cyclin-dependent kinase; **ctDNA**, circulating tumor DNA; **ECOG**, Eastern Cooperative Oncology Group.

Table 2. Patient disposition

	OP-1250 dose				
Treatment status	30 mg (n=3)	60 mg (n=3)	90 mg (n=3)	120 mg (n=20)	Total (N=29)
Treatment ongoing	1	2	2	12	17 (59)
Treatment discontinued	2	1	1	8	12 (41)
Reasons for treatment discontinuation					
Physician decision	0	0	0	1	1 (3)
Disease progression	2	1	1	6	10 (34)
Patient decision	0	0	0	1	1 (3)

Data shown are n or n (%).

Safety and Tolerability

- No DLTs were observed during the dose-escalation part of the study
- As of the data cutoff date, most TEAEs were grade 1 or 2 (**Table 3**)
- There were no dose-related increases in incidence or severity of TEAEs
- There were no treatment-related fatal events
- No patients discontinued treatment due to a TEAE
- The most common TEAEs (>20% of patients) included neutropenia, gastrointestinal events (nausea, vomiting, constipation, diarrhea, gastroesophageal reflux disease), and thrombocytopenia (**Table 3**)

Table 3. Treatment-emergent adverse events (TEAEs) reported in ≥20% of patients

	OP-1250 dose									
TEAE	30 mg (n=3)		60 mg (n=3)		90 mg (n=3)		120 mg (n=20)		Total (N=29)	
	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3
Neutropenia	2	2	3	2	3	2	11	10	19 (66)	16 (55)
Nausea	2	0	2	0	1	0	9	0	14 (48)	0
Vomiting	1	0	2	0	1	0	4	0	8 (28)	0
Constipation	1	0	1	0	0	0	4	0	6 (21)	0
Diarrhea	1	0	1	0	0	0	4	0	6 (21)	0
GERD	2	0	1	0	0	0	3	0	6 (21)	0
Thrombocytopenia	0	0	1	0	0	0	5	0	6 (21)	0

Data shown are n or n (%).
GERD, gastroesophageal reflux disease.

- There were five serious adverse events: grade 2 malignant pleural effusion (n=1), grade 3 colitis (n=1), grade 4 neutropenia (n=2), and grade 5 multiple organ dysfunction syndrome (n=1). All were considered by the investigator as not related to OP-1250, except neutropenia in one patient that was considered related to OP-1250 and palbociclib
- Grade 3 or 4 neutropenia was reported in 16 (55%) patients
 - 14 patients (48%) had grade 3 neutropenia, all occurring during the first treatment cycle and assessed as related to palbociclib in all patients and possibly related to OP-1250 in 6 patients
 - 2 patients (7%) had grade 4 neutropenia, both starting at week 4; in one patient neutropenia was assessed as related to palbociclib and unrelated to OP-1250, in the other patient it was assessed as related to palbociclib and possibly related to OP-1250
 - 6 patients had dose reductions of palbociclib due to neutropenia. No patients had dose reductions of OP-1250 for neutropenia
 - Neutropenia was reversible in all patients and the timing was consistent with palbociclib-related neutropenia

Pharmacokinetics

- There was no DDI between palbociclib and OP-1250 in the 30–120 mg dose range
 - OP-1250 did not affect palbociclib 125 mg exposure when compared with published concentrations for single-agent palbociclib[8] (**Figure 3**)
 - The geometric mean parameters of palbociclib for each of the combination dose levels were within the geometric mean ± GeoSD of the reference data
 - Palbociclib did not affect OP-1250 PK; the rate and extent of absorption and half-life were consistent with single agent OP-1250 (**Figure 4**; **Table 4**)
- OP-1250 was readily bioavailable and showed dose-proportional exposure and a long half-life
- Steady state plasma levels showed minimal peak-to-trough variability, enabling consistent steady state target coverage

Pharmacokinetics, cont'd

Figure 3. Steady state exposure ($AUC_{0–24}$ and C_{max}) of palbociclib (125 mg) alone and in combination with OP-1250[a]

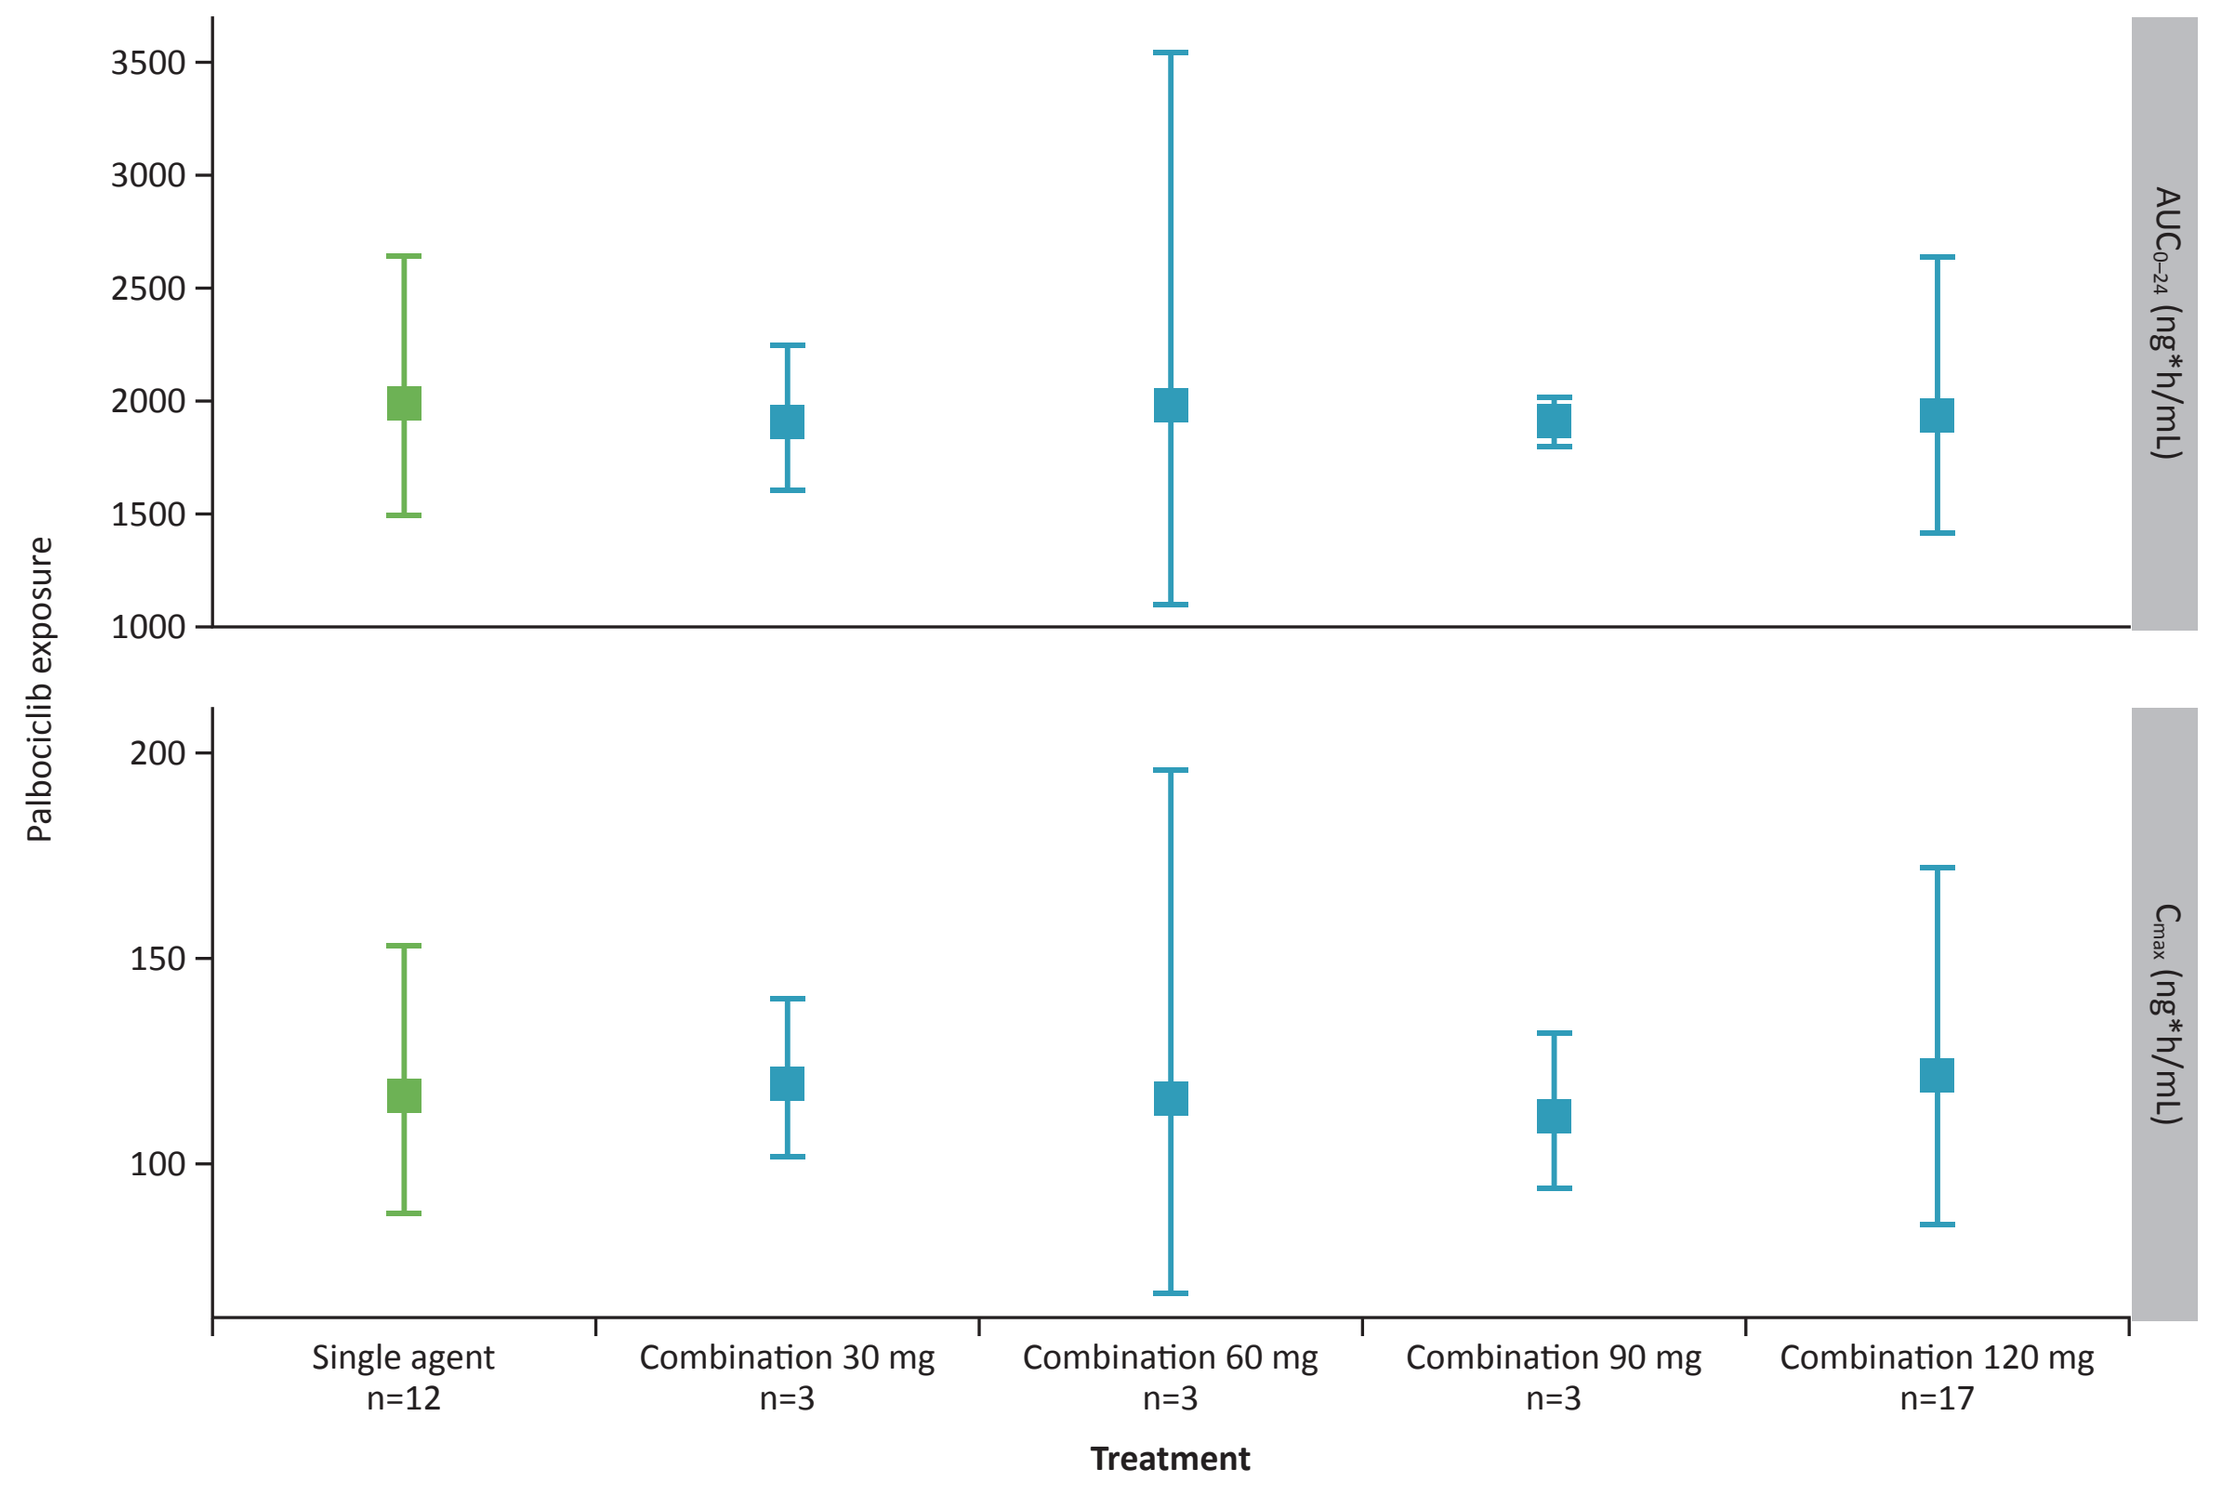
3500
3000
2500
2000
1500
1000
200
150
100
Palbociclib exposure
AUC0–24 (ng*h/mL)
Cmax (ng*h/mL)
Single agent n=12
Combination 30 mg n=3
Combination 60 mg n=3
Combination 90 mg n=3
Combination 120 mg n=17
Treatment

Note: data are GeoMean±GeoSD.
[a]OP-1250 did not affect steady state palbociclib exposure when compared with published exposures for single-agent palbociclib[8]
$AUC_{0–24}$, area under the concentration time curve from 0 to 24 h; **C_{max}**, maximum concentration.

Table 4. OP-1250 pharmacokinetics in combination with palbociclib (125 mg) at steady state

OP-1250 dose	C_{min} (ng/mL)	C_{max} (ng/mL)	C_{avg} (ng/mL)	T_{max} (h)	$AUC_{0–24}$ (ng*h/mL)
30 mg (n=2)	61.2 (38%)	136 (49%)	94.7 (13%)	2.0 (2.0–2.0)	2270 (13%)
60 mg (n=3)	186 (52%)	337 (24%)	253 (43%)	2.0 (2.0–2.0)	6070 (43%)
90 mg (n=3)	199 (55%)	549 (63%)	341 (49%)	2.0 (2.0–4.0)	8180 (49%)
120 mg (n=12)	313 (43%)	573 (43%)	429 (39%)	4.0 (2.0–6.0)	10300 (39%)

Data shown are GeoMean(GeoCV%) or median (min–max). Plasma samples for OP-1250 assessment were collected on C1 day (D) 1 (C1D1), C1D2/D8/D15, C2D15/D16, C3D1/D15, C5D1, C7D1, and C9D1. For palbociclib, they were collected on C1D15, C2D15, and C3D15.
AUC_{0-24}, area under the concentration time curve from 0 to 24 h; **C_{avg}**, average concentration; **C_{max}**, maximum concentration; **C_{min}**, minimum concentration; **max**, maximum; **min**, minimum; **PK**, pharmacokinetics; **T_{max}**, time to maximum concentration.

Figure 4. Steady state exposure ($AUC_{0–24}$ and C_{max}) of OP-1250 alone and in combination with palbociclib (125 mg)[a]

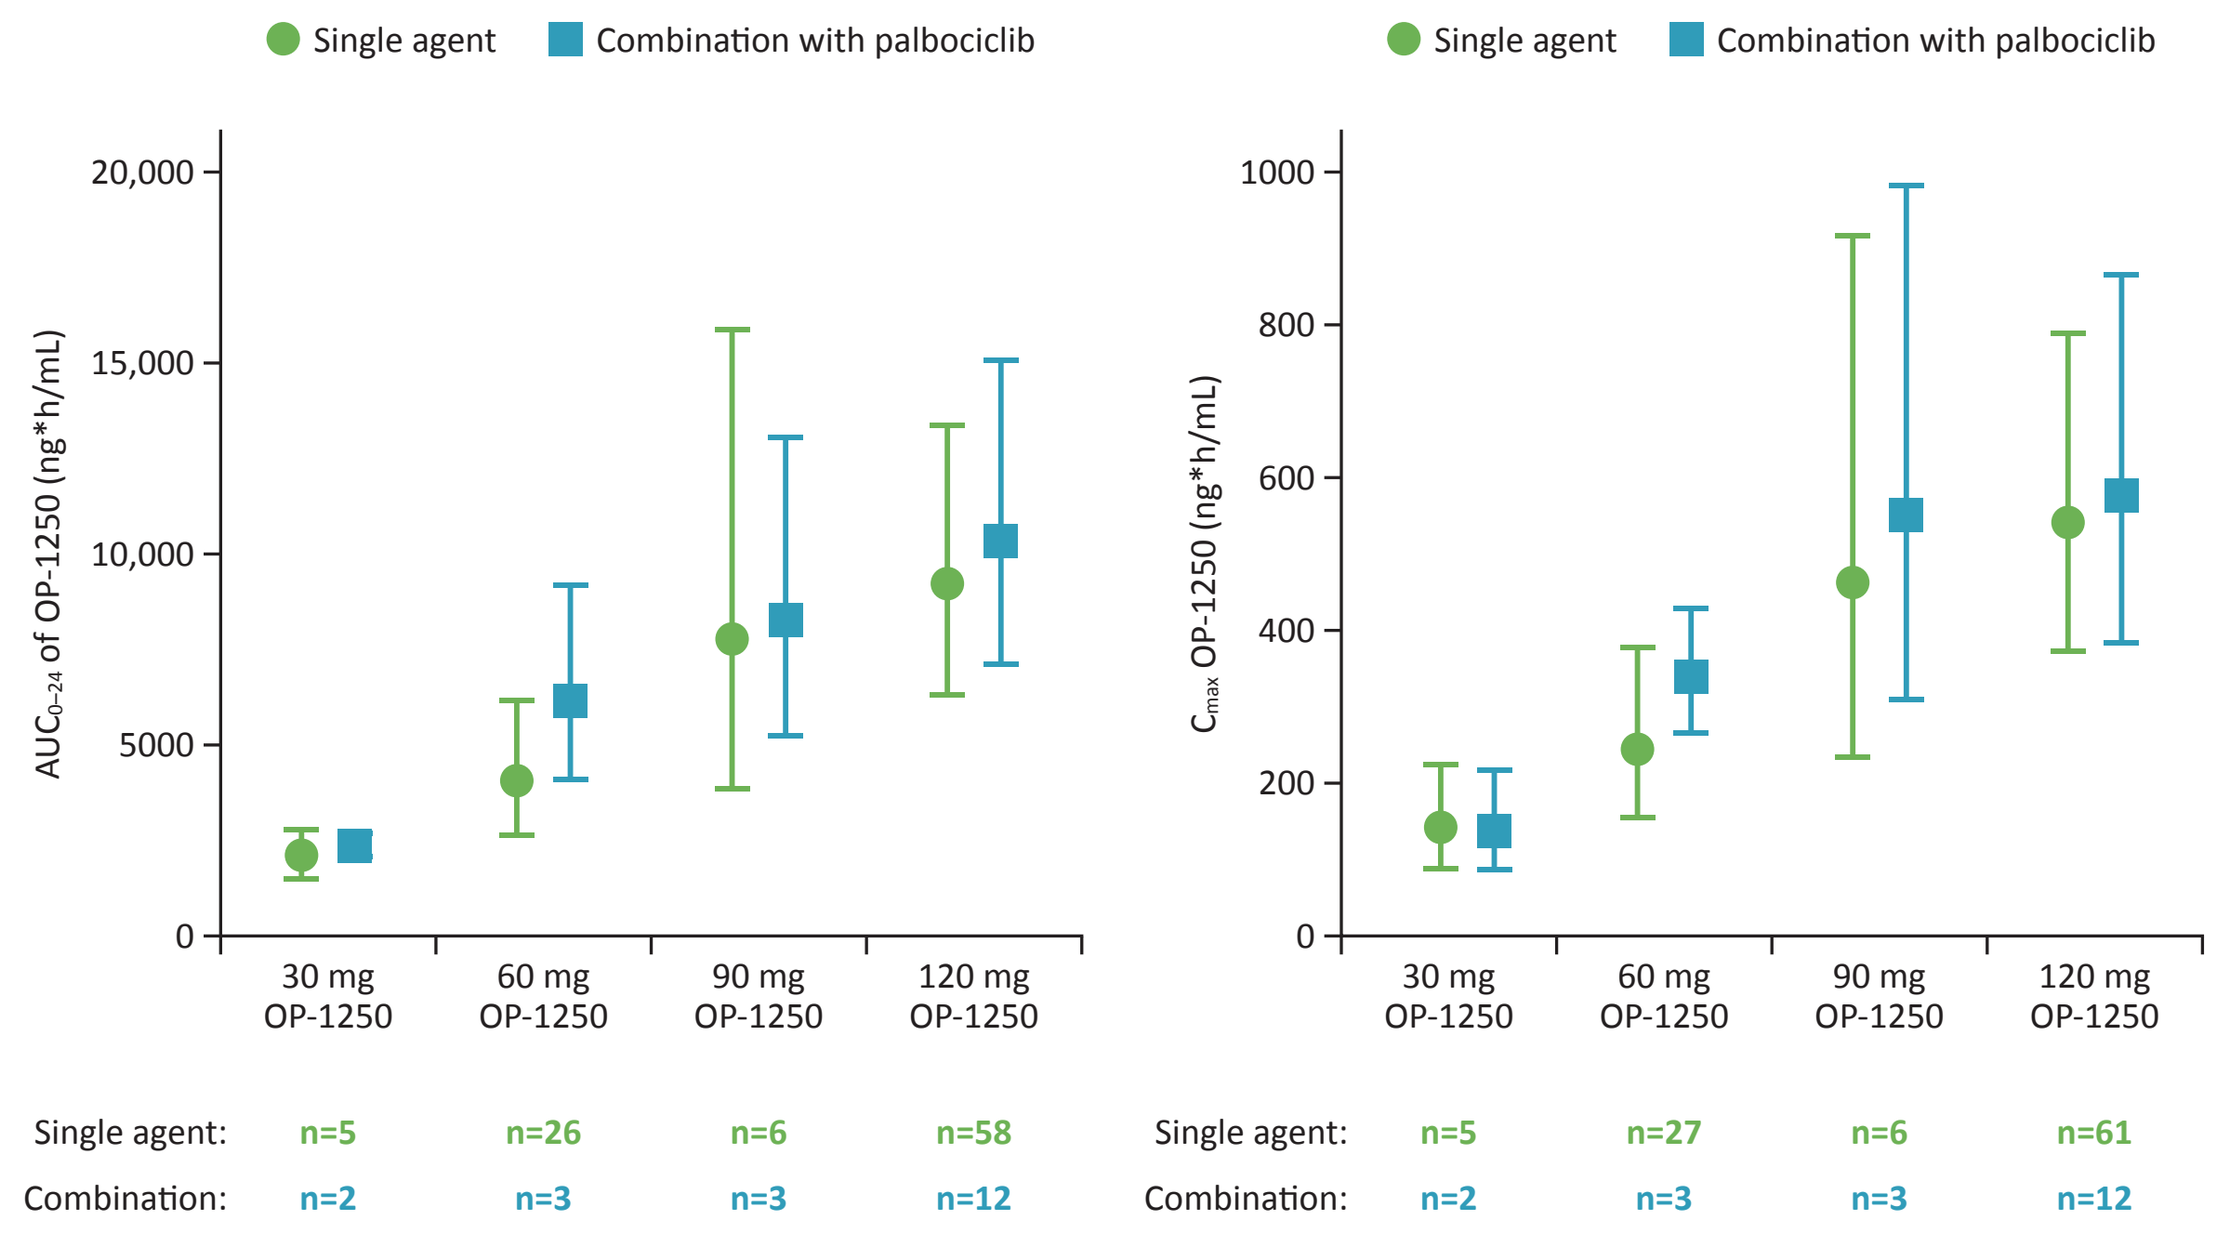
Single agent
Combination with palbociclib
20,000
15,000
10,000
5000
0
AUC0–24 of OP-1250 (ng*h/mL)
1000
800
600
400
200
0
Cmax OP-1250 (ng*h/mL)
30 mg OP-1250
60 mg OP-1250
90 mg OP-1250
120 mg OP-1250
Single agent: n=5 n=26 n=6 n=58
Combination: n=2 n=3 n=3 n=12
Single agent: n=5 n=27 n=6 n=61
Combination: n=2 n=3 n=3 n=12

Note: data are GeoMean±GeoSD.
[a]Palbociclib did not affect steady state OP-1250 exposure compared with OP-1250 single-agent exposure seen in an ongoing monotherapy trial.
$AUC_{0–24}$, area under the concentration time curve from 0 to 24 h; **C_{max}**, maximum concentration.

Antitumor Activity

Figure 5. Duration of treatment[a]

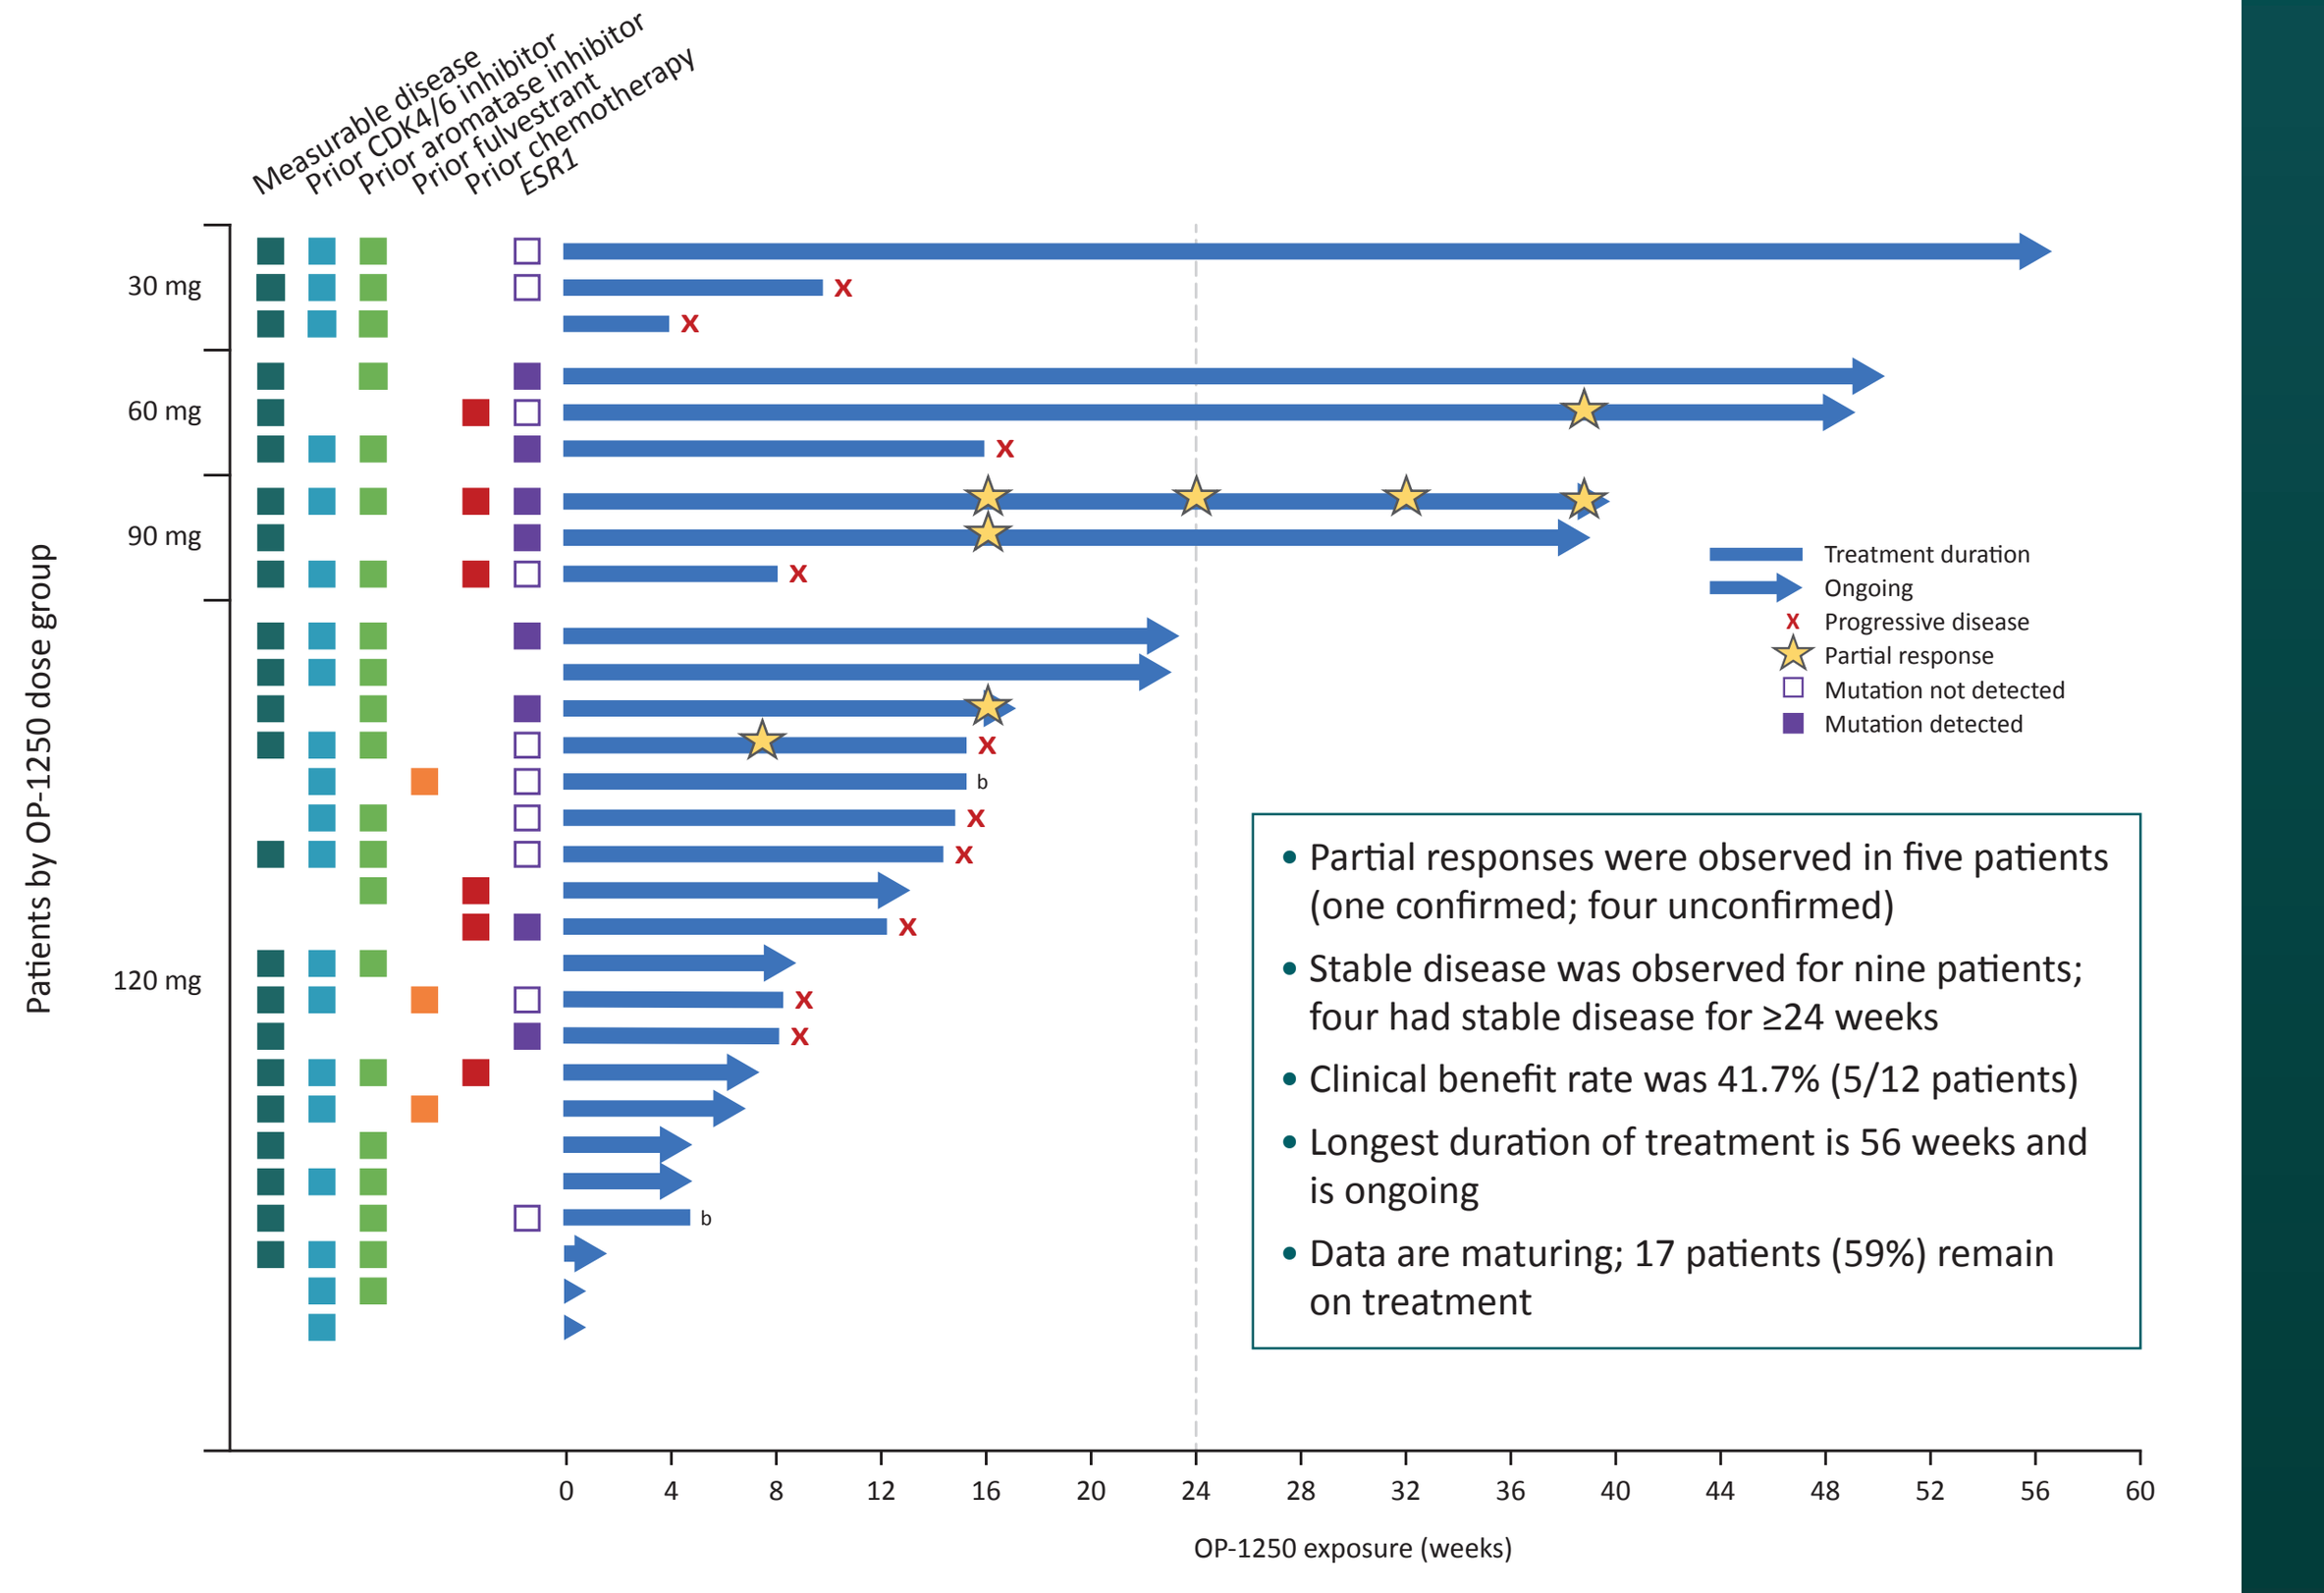
Measurable disease
Prior CDK4/6 inhibitor
Prior aromatase inhibitor
Prior fulvestrant
Prior chemotherapy
ESR1
30 mg
60 mg
90 mg
120 mg
Patients by OP-1250 dose group
Treatment duration
Ongoing
Progressive disease
Partial response
Mutation not detected
Mutation detected
• Partial responses were observed in five patients (one confirmed; four unconfirmed)
• Stable disease was observed for nine patients; four had stable disease for ≥24 weeks
• Clinical benefit rate was 41.7% (5/12 patients)
• Longest duration of treatment is 56 weeks and is ongoing
• Data are maturing; 17 patients (59%) remain on treatment
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60
OP-1250 exposure (weeks)

[a]Each lane represents one study patient. [b]Two patients discontinued treatment due to patient's decision or physician's decision.
CDK, cyclin-dependent kinase.

Conclusions

- The combination of OP-1250 as 30–120 mg with palbociclib 125 mg was safe and well tolerated; no new safety signals were identified
 - There were no dose-related increases in the incidence, severity, or timing of adverse events
 - The incidence and severity of adverse events were consistent with the expected safety profile of palbociclib plus endocrine therapy and were similar (including neutropenia) to those reported with palbociclib plus letrozole in the PALOMA 2 study[9] which demonstrated all grade neutropenia, 80%; grade 3 neutropenia, 56%; grade 4 neutropenia, 10%
- Tumor responses and prolonged disease stabilization were observed in this group of patients, including in those previously exposed to CDK4/6 inhibitors
- No DDI between OP-1250 and palbociclib was observed
 - There was no induced metabolism or increase in exposure of palbociclib when administered in combination with OP-1250
 - When administered in combination with palbociclib, OP-1250 exposure was consistent with the observed exposure of OP-1250 monotherapy
- Findings from this study support the ongoing clinical development of OP-1250 in combination with CDK4/6 inhibitors for the treatment of HR+/HER2– MBC

References

1. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Breast Cancer. V2.2023. **2.** Rasha F, et al. *Mol Cell Endocrinol.* 2021;532:111322. **3.** Alemany C, et al. Presented at AACR-NCI-EORTC. October 7–10, 2021. Abstract number P037. **4.** Shang Y, Brown M. *Science.* 2002;295:2465–8. **5.** Webb P, et al. *J Biol Chem.* 2003;278:6912–20. **6.** Hamilton E, et al. Presented at EORTC–NCI–AACR Molecular Targets and Cancer Therapeutics Symposium. October 26–29, 2022. Poster number 101/PB091. **7.** Chan A, et al. Presented at San Antonio Breast Cancer Conference. Dec 6–10, 2022. Poster number 101/PB091. **8.** Pfizer Inc. Pfizer Canada ULC. 2021. Available at: https://www.pfizer.ca/files/Ibrance_PM_EN_243405_15-Jul-2021.pdf. (Accessed March 30, 2023). **9.** Pfizer Inc. Ibrance (Palbociclib) prescribing information. 2019.

Acknowledgments

We thank the OP-1250 trial patients, their families and caregivers, trial investigators, and study staff. This study was sponsored by Olema Oncology. Editorial support was provided by Twist Medical, LLC, and funded by Olema Oncology.

Disclosures

Dr. Chan has served on advisory boards for Specialised Therapeutics, has an unrestricted research grant from Eisai, and has received honoraria as an invited speaker for Pierre Fabre and Novartis.

Presented at the ESMO Breast Cancer Annual Congress; May 11–13, 2023; Berlin, Germany

Sponsored by Olema Oncology